Antisense oligonucleotide-based strategy to target hepatitis delta virus infections

Yannick Debing[1], Caroline Pons[2], Faustine Bernardin[2], Lars Degrauwe[1], Kellan Passow[3], Isabel Heremans[1], Aneerban Bhattacharya[3], Antitsa Stoycheva[3], Lawrence Blatt[4], Julian A. Symons[3], Andreas Jekle[3], Vivek Rajwanshi[3], David B. Smith[3], Eloi R. Verrier[2], David Durantel[2] and Julie Lucifora[2]

[1]Aligos Belgium BV, Gaston Geenslaan 1, 3001 Leuven, Belgium

[2]CIRI, Centre International de Recherche en Infectiologie, Univ Lyon, Inserm, U1111, Université Claude Bernard Lyon 1, CNRS, UMR5308, ENS de Lyon, F-69007, Lyon, France

[3]Aligos Therapeutics, Inc., 1 Corporate Drive, South San Francisco, CA 94080

Contact information

julie.lucifora@inserm.fr

ydebing@aligos.com

EASL CONGRESS Barcelona Spain 27–30 May 2026

Background and aims

Currently approved treatments for chronic hepatitis delta virus (HDV) infection (Peg-IFN-α and bulevirtide) rarely cure patients in a finite treatment regimen. New direct-acting agents (DAA), targeting HDV replication, could therefore be a valuable addition to the current therapeutic arsenal.

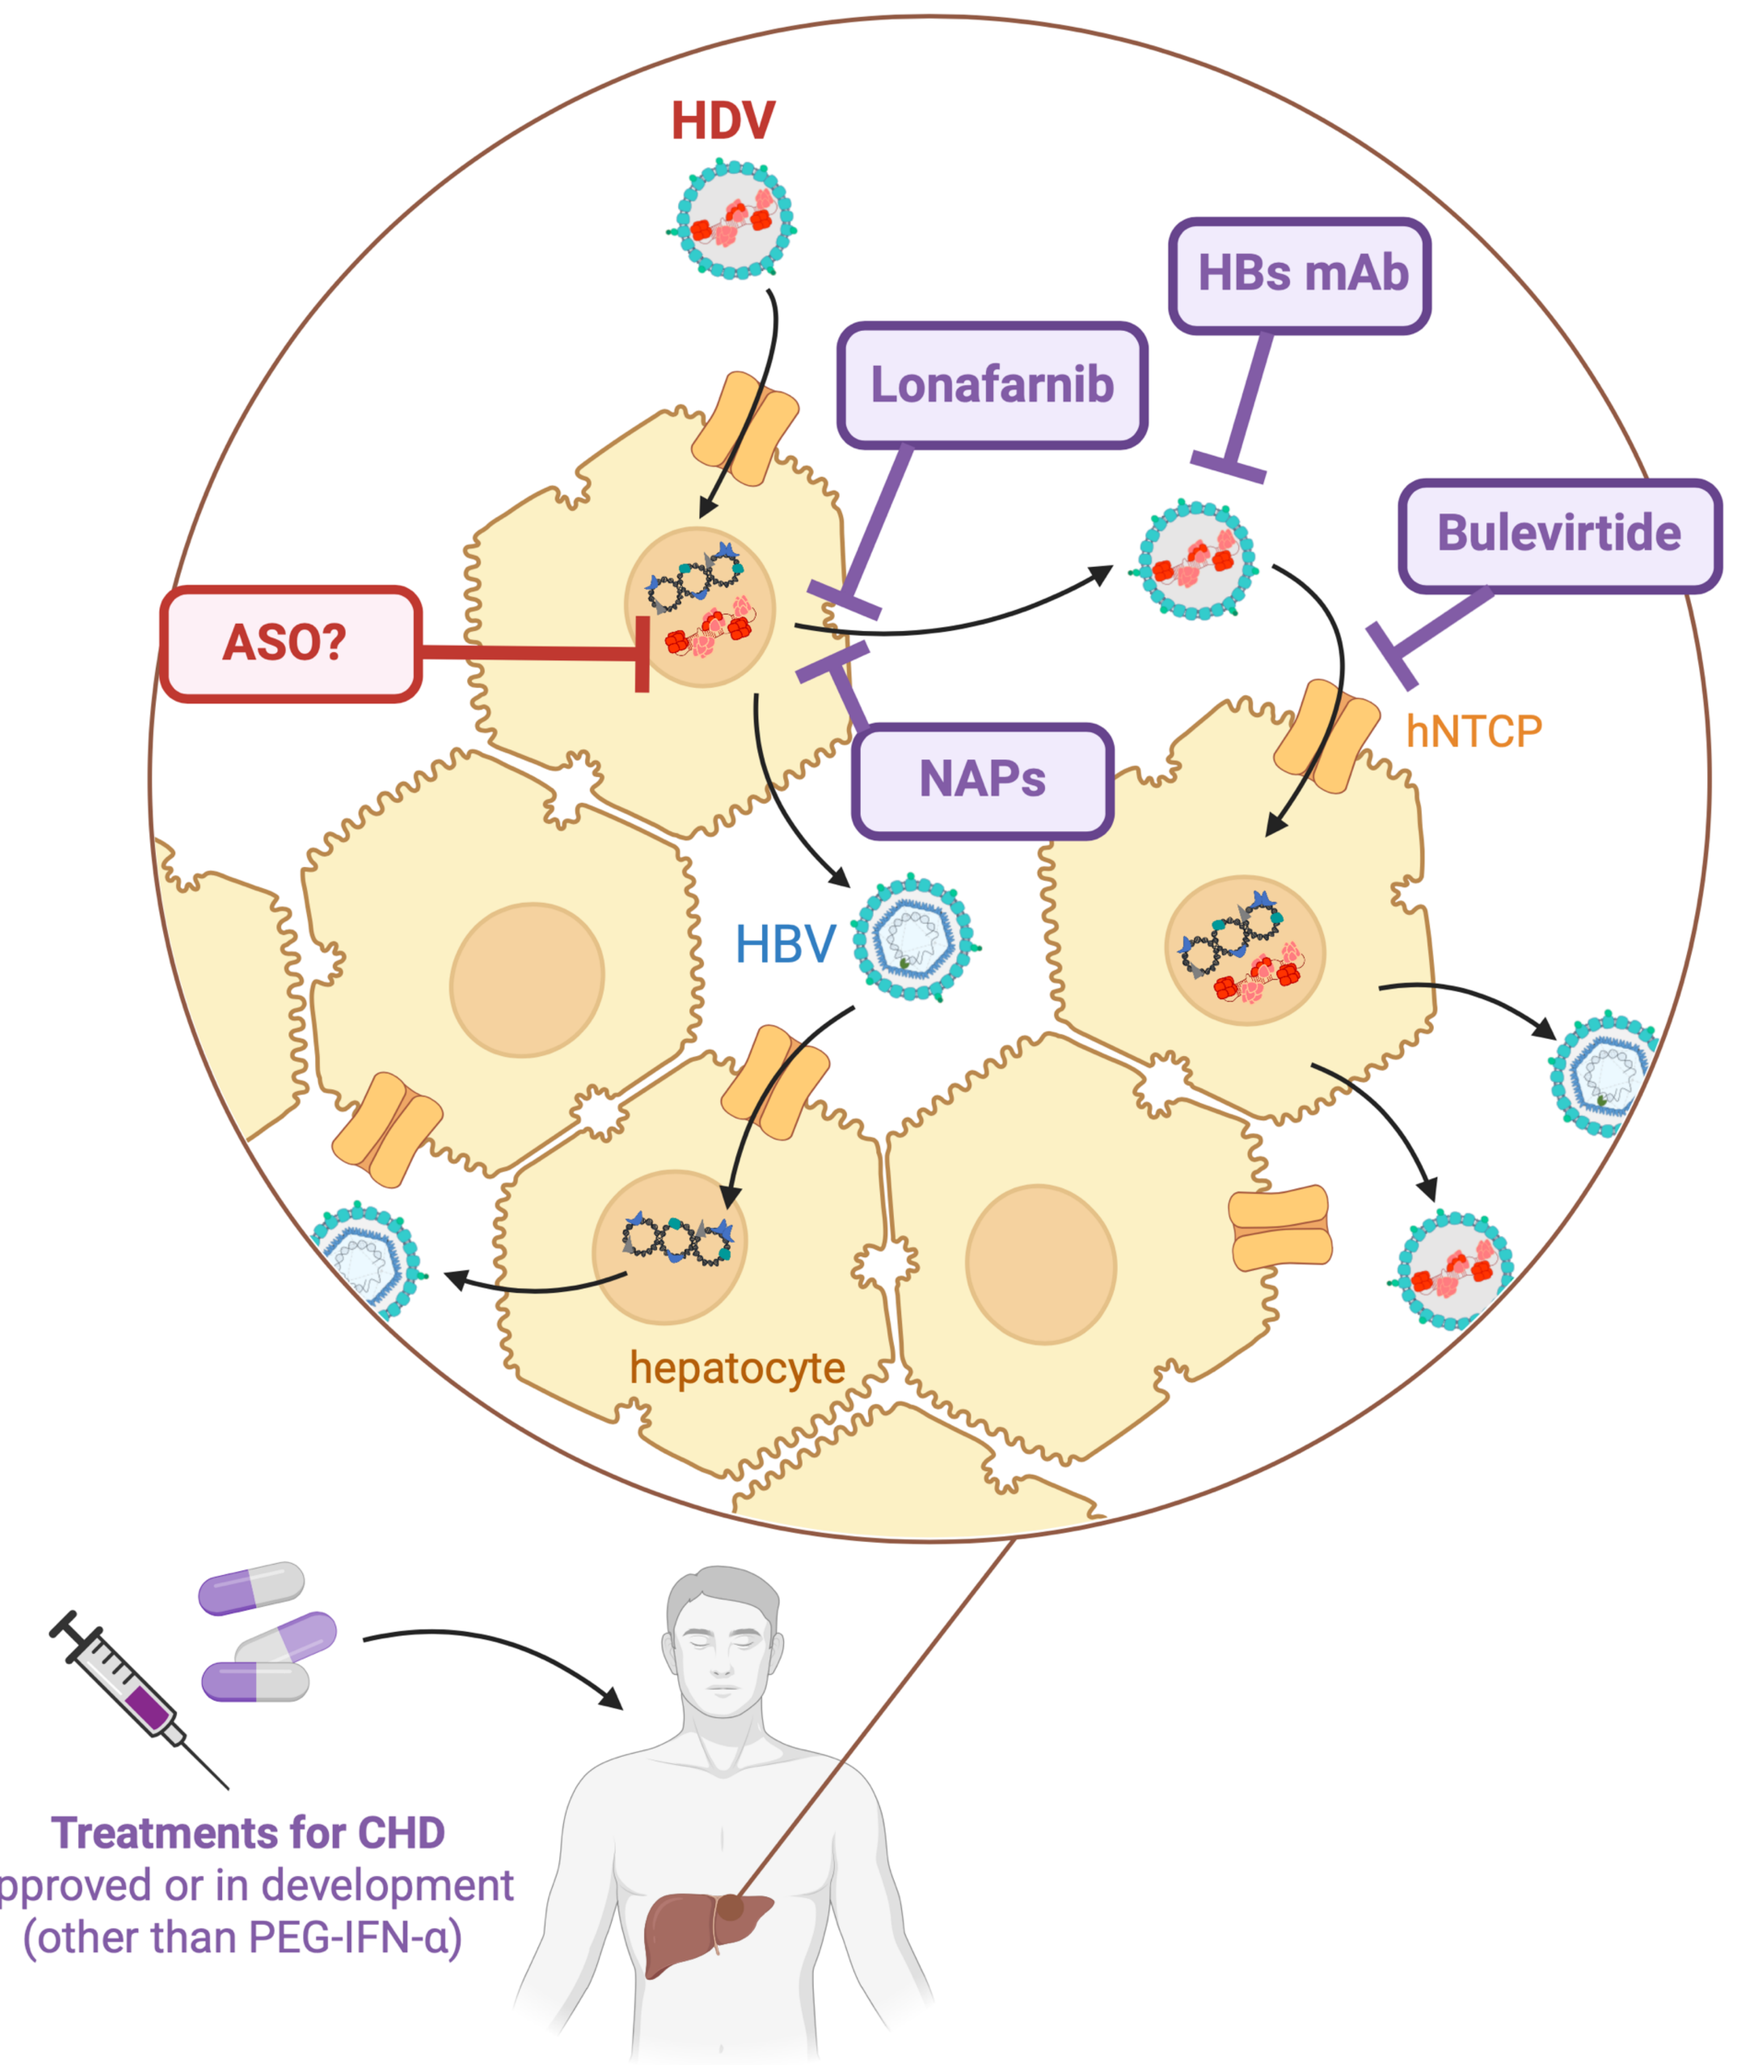
HDV
HBs mAb
Lonafarnib
Bulevirtide
ASO?
NAPs
hNTCP
HBV
hepatocyte
Treatments for CHD
approved or in development
(other than PEG-IFN-α)

Since HDV is an RNA virus replicating its genome via a rolling-circle process in the nucleus, and antisense oligonucleotides (ASOs) are known to act both in the cytoplasm and the nucleus, we hypothesized that ASOs targeting HDV RNAs may potently and specifically inhibit intracellular HDV RNA amplification irrespective of helper HBV. We synthesized 112 different 5-10-5 gapmer ASOs targeting conserved regions of HDV and screened them for their antiviral effect on HDV. The HBV-directed ASO, bepirovirsen, was used as a non-targeting ASO control.

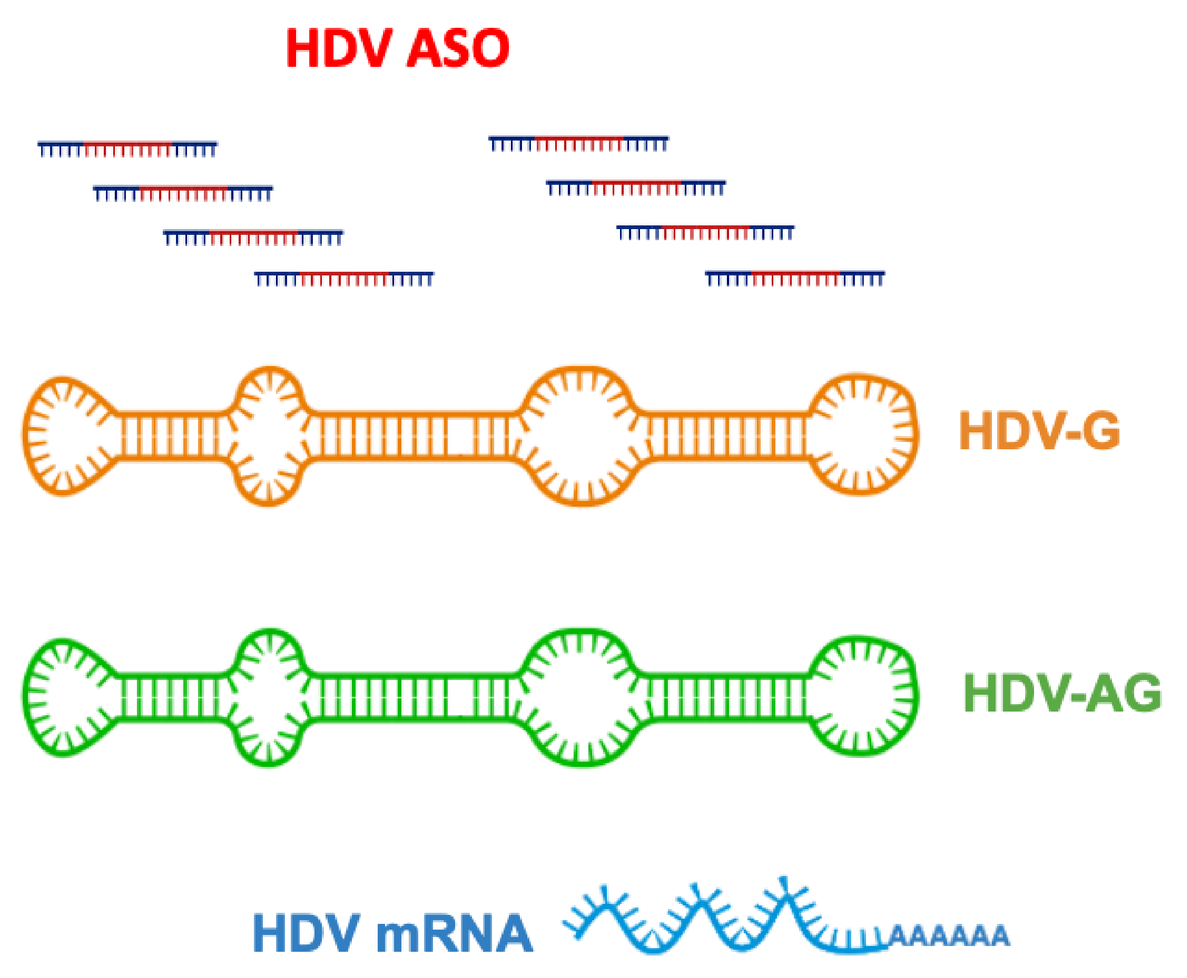
HDV ASO
HDV-G
HDV-AG
HDV mRNA

1. Identification of anti-HDV ASOs with antiviral effects

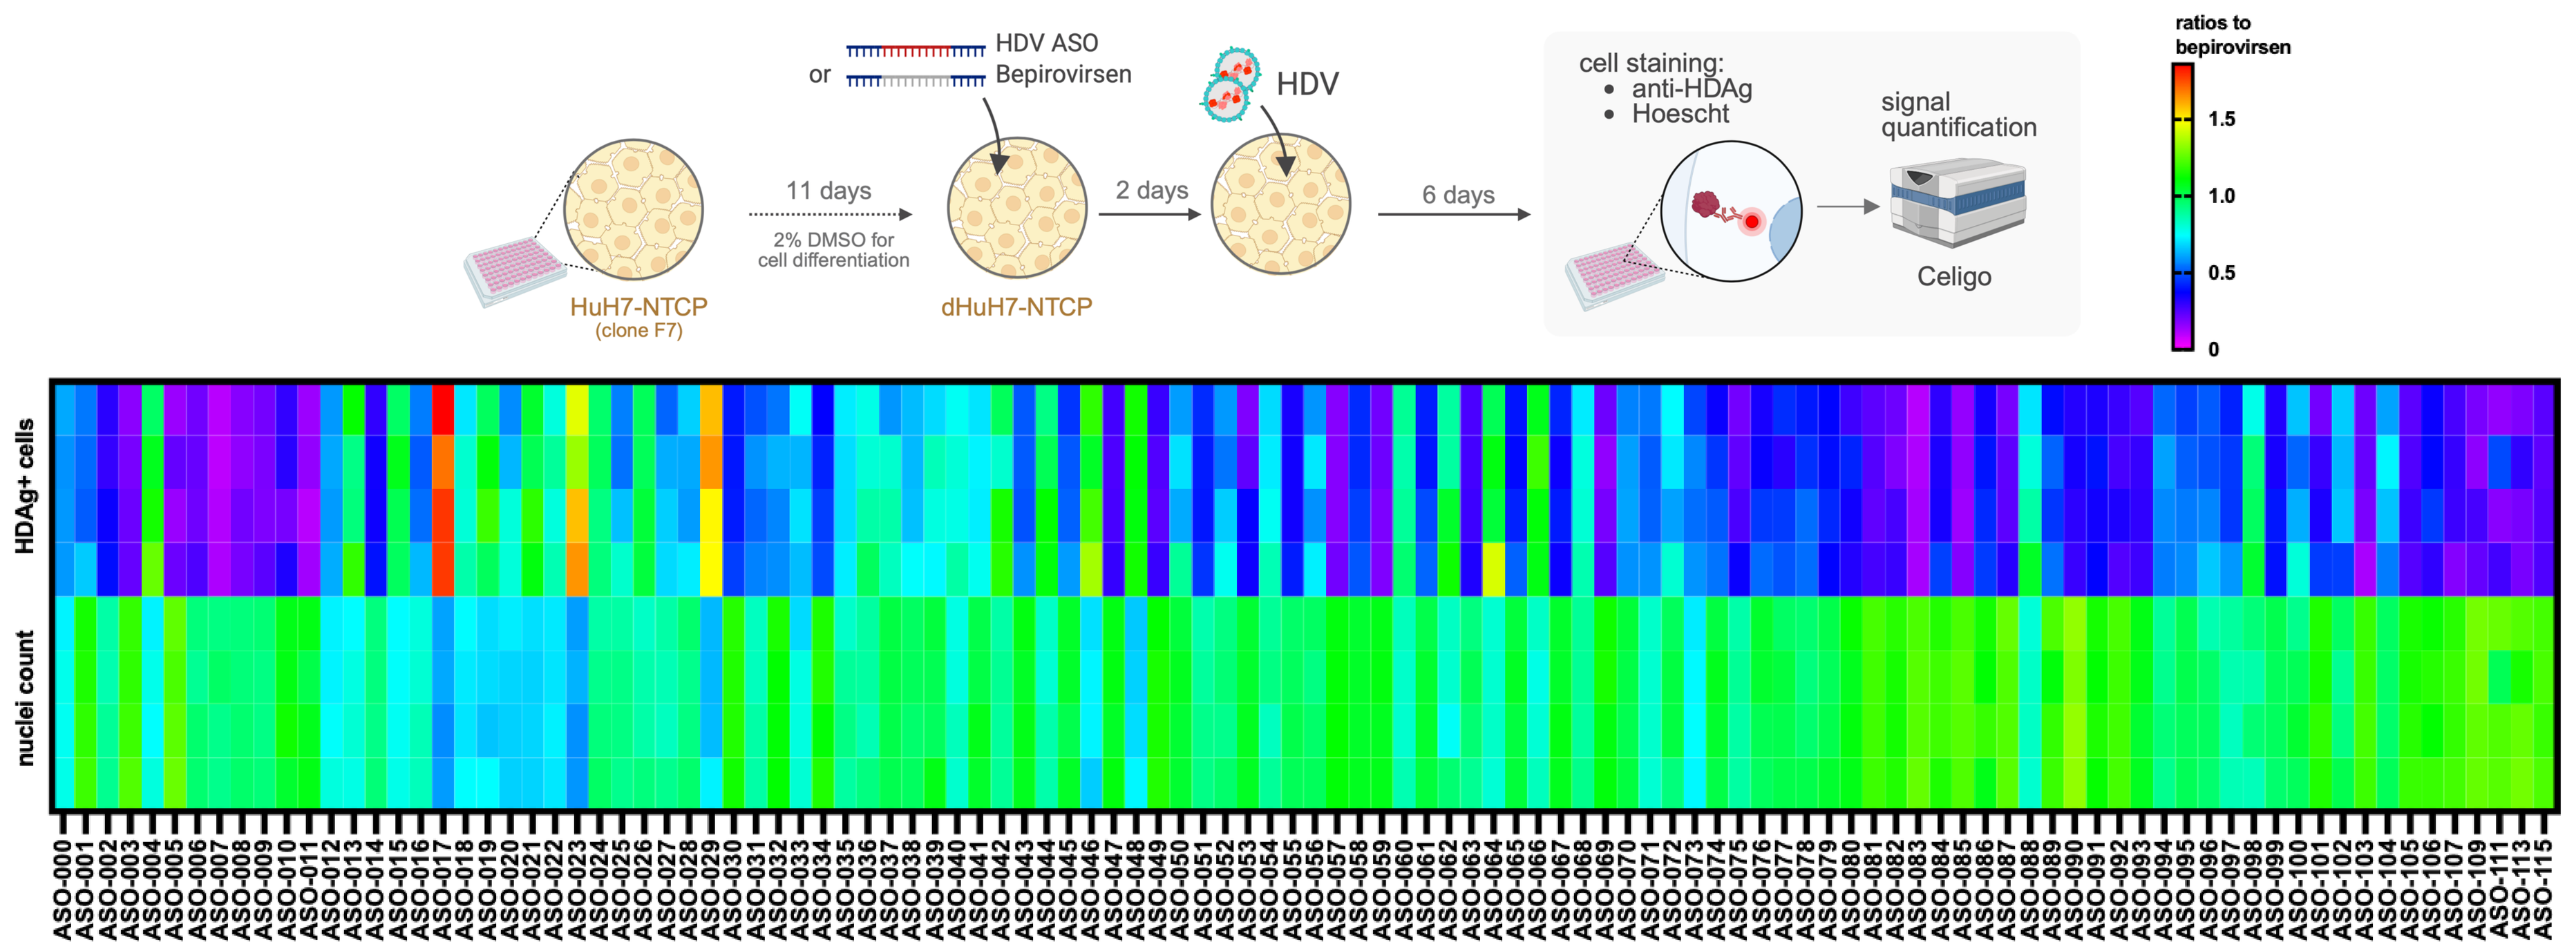
HDV ASO
or Bepirovirsen
HDV
11 days
2% DMSO for cell differentiation
2 days
6 days
HuH7-NTCP (clone F7)
dHuH7-NTCP
cell staining:
anti-HDAg
Hoescht
signal quantification
Celigo
ratios to bepirovirsen
HDAg+ cells
nuclei count

➢ 19 anti-HDV ASO candidates allowed a ≥ 75 % reduction of the levels of HDAg+ cells without toxicity

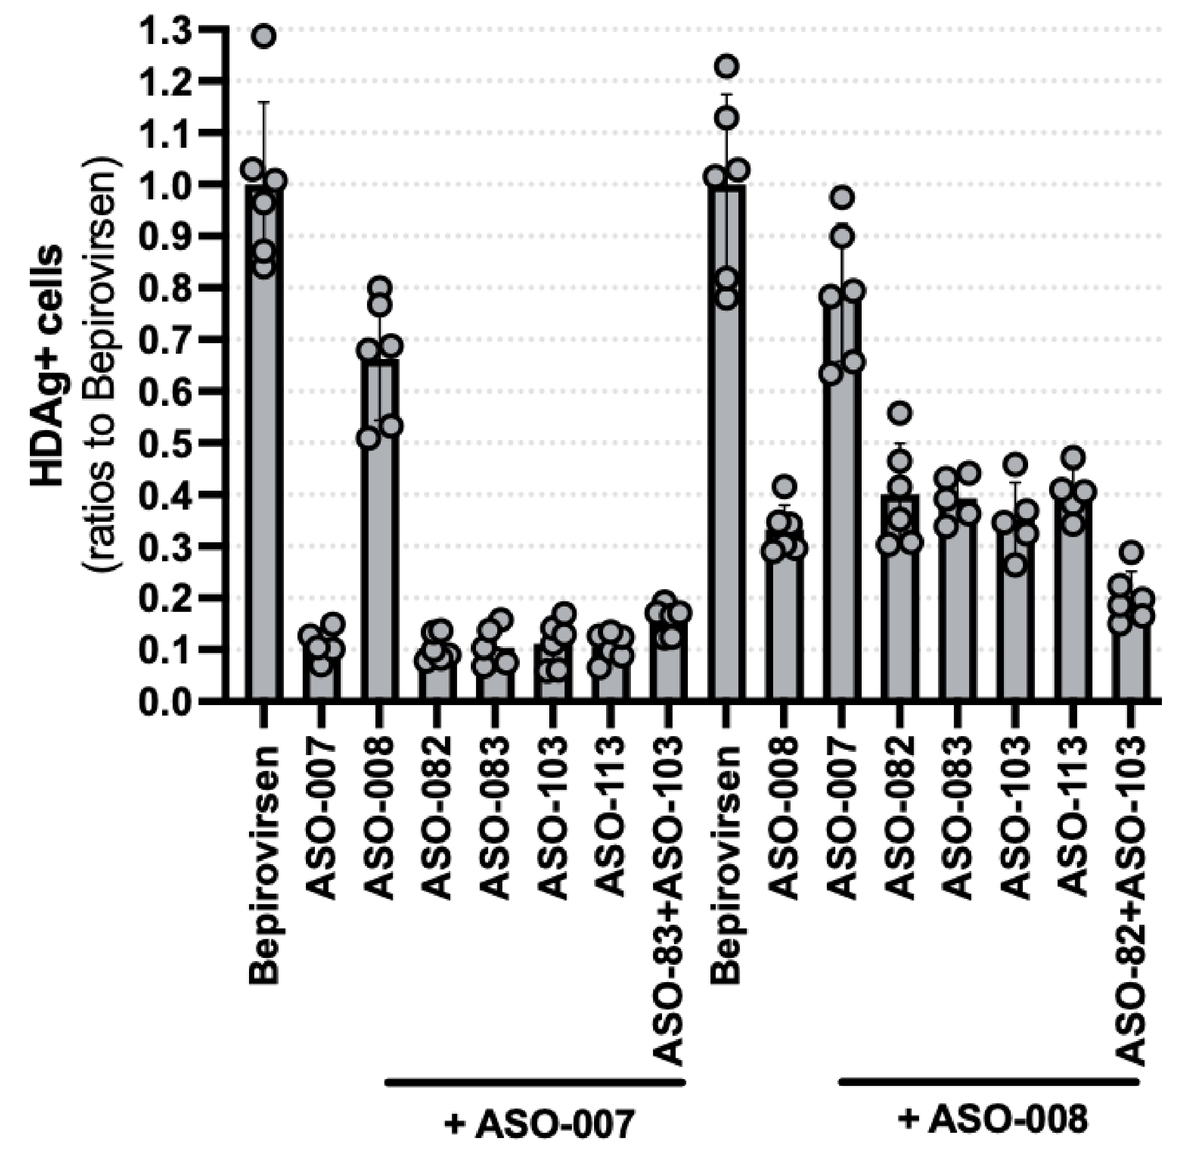
HDAg+ cells (ratios to Bepirovirsen)
Bepirovirsen
ASO-007
ASO-008
ASO-082
ASO-083
ASO-103
ASO-113
ASO-83+ASO-103
ASO-82+ASO-103
+ ASO-007
+ ASO-008

➢ Combination of active anti-HDV ASOs did not increase the antiviral effects and complementary ASO (007 and 008) are antagonists

2. Anti-HDV ASO ALG-215007 decreases all HDV replication markers

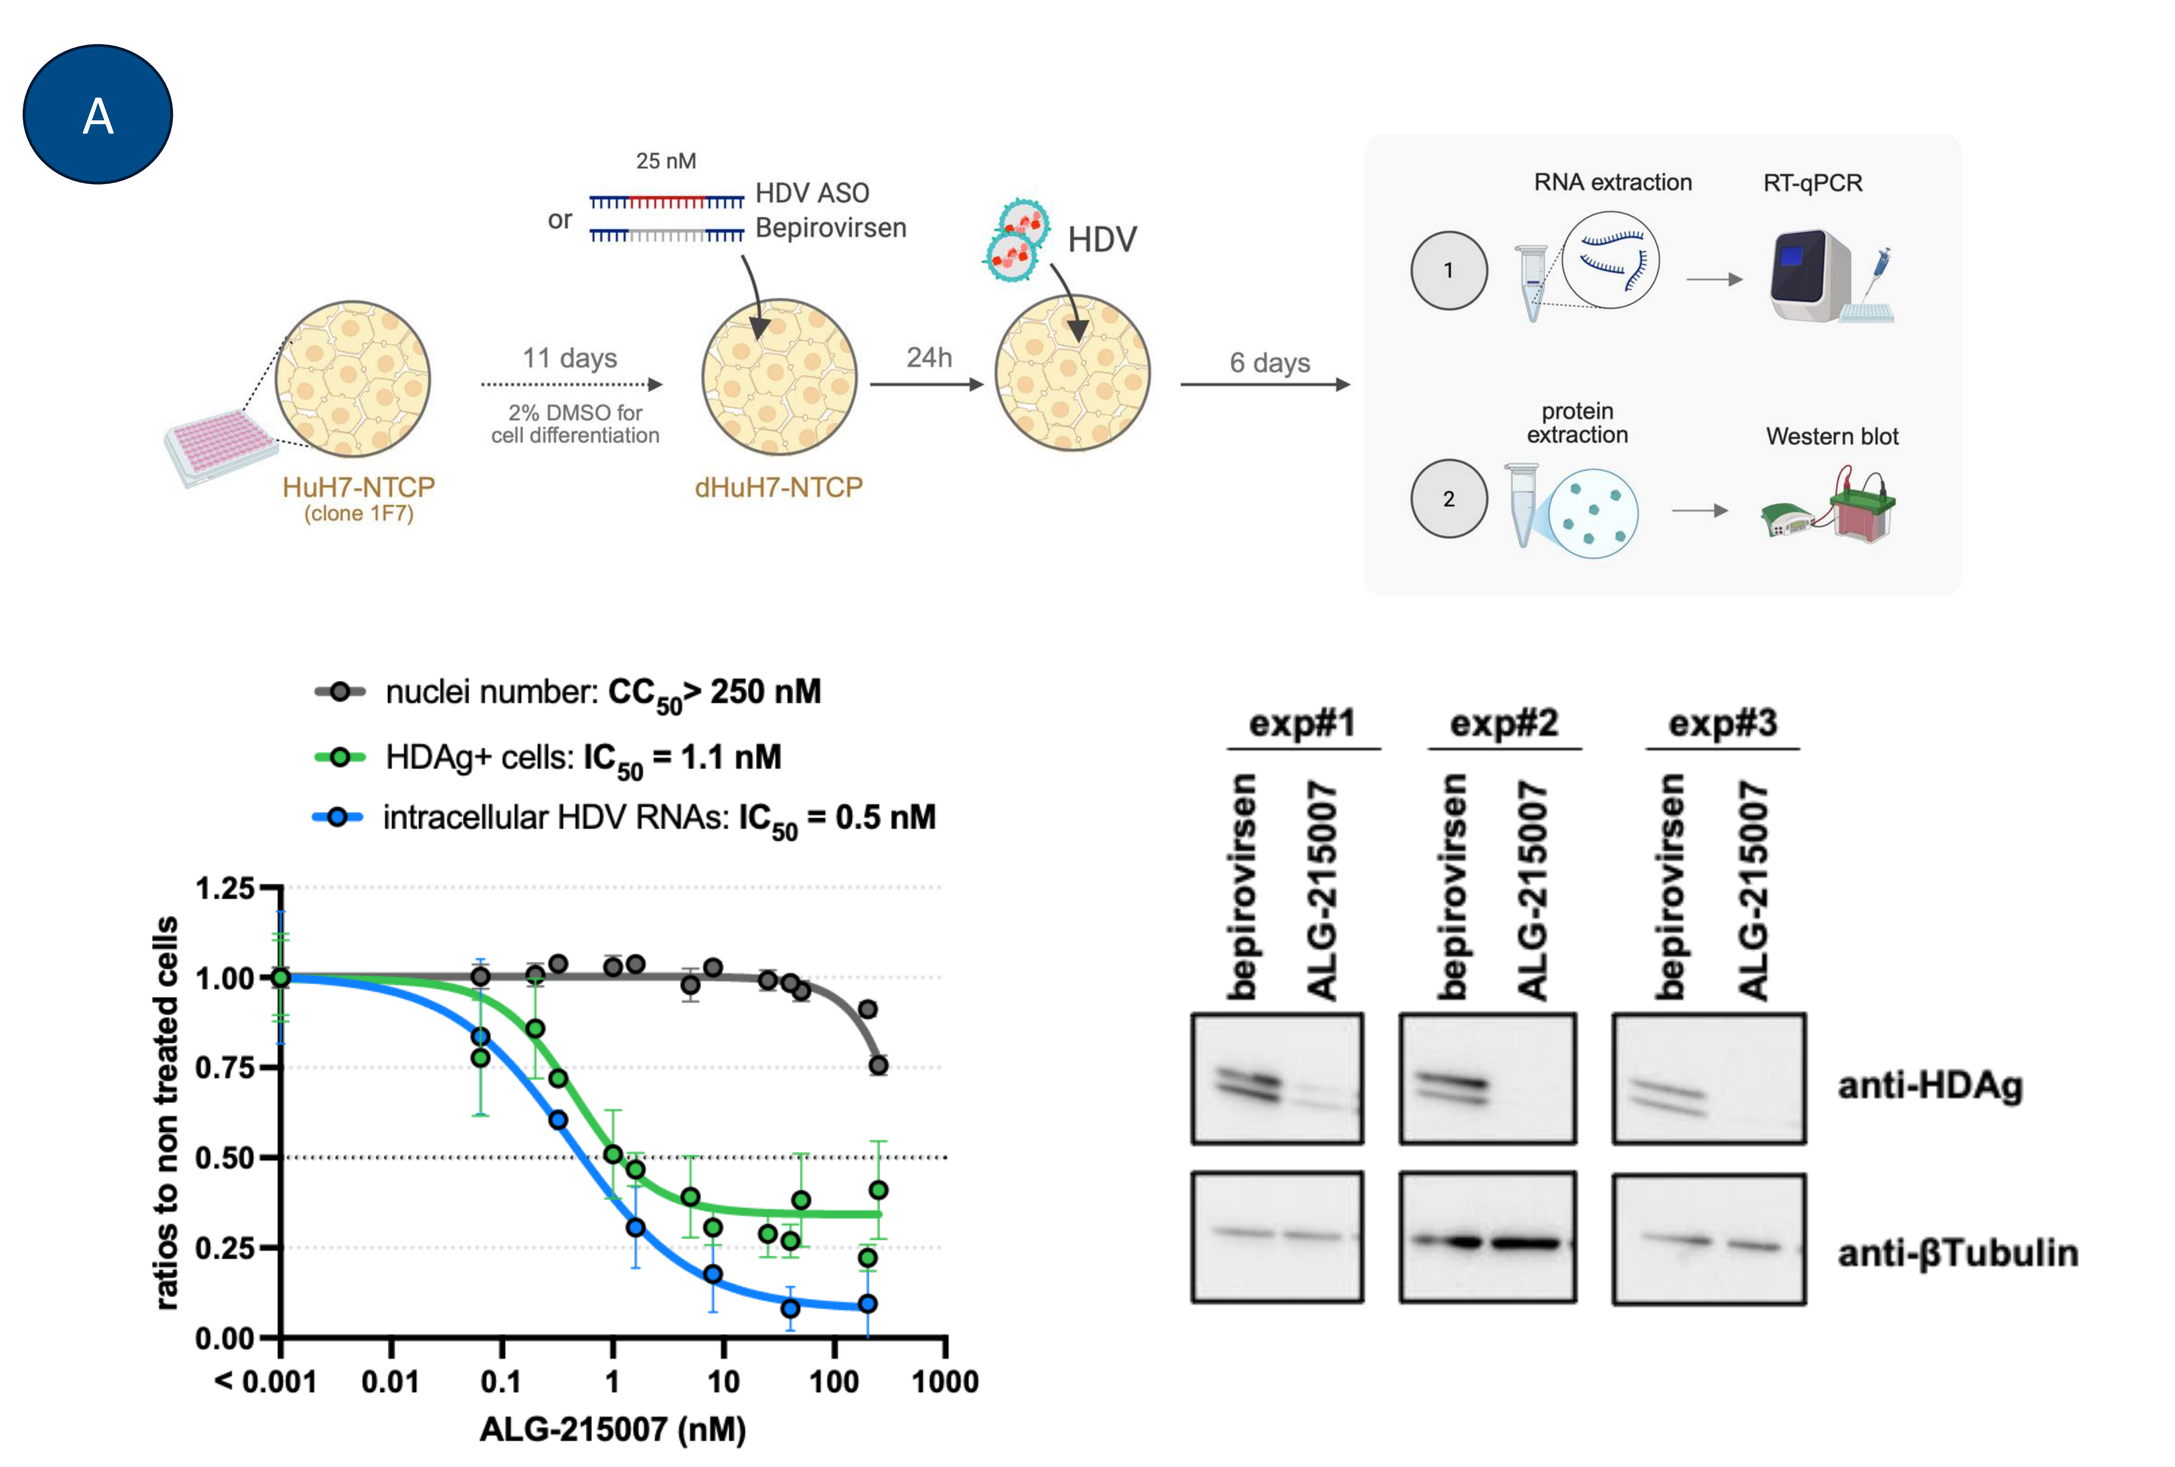
A
25 nM
HDV ASO
or Bepirovirsen
HDV
HuH7-NTCP (clone 1F7)
11 days
2% DMSO for cell differentiation
dHuH7-NTCP
24h
6 days
RNA extraction
RT-qPCR
protein extraction
Western blot
nuclei number: CC50 > 250 nM
HDAg+ cells: IC50 = 1.1 nM
intracellular HDV RNAs: IC50 = 0.5 nM
ratios to non treated cells
ALG-215007 (nM)
exp#1
exp#2
exp#3
bepirovirsen
ALG-215007
anti-HDAg
anti-βTubulin

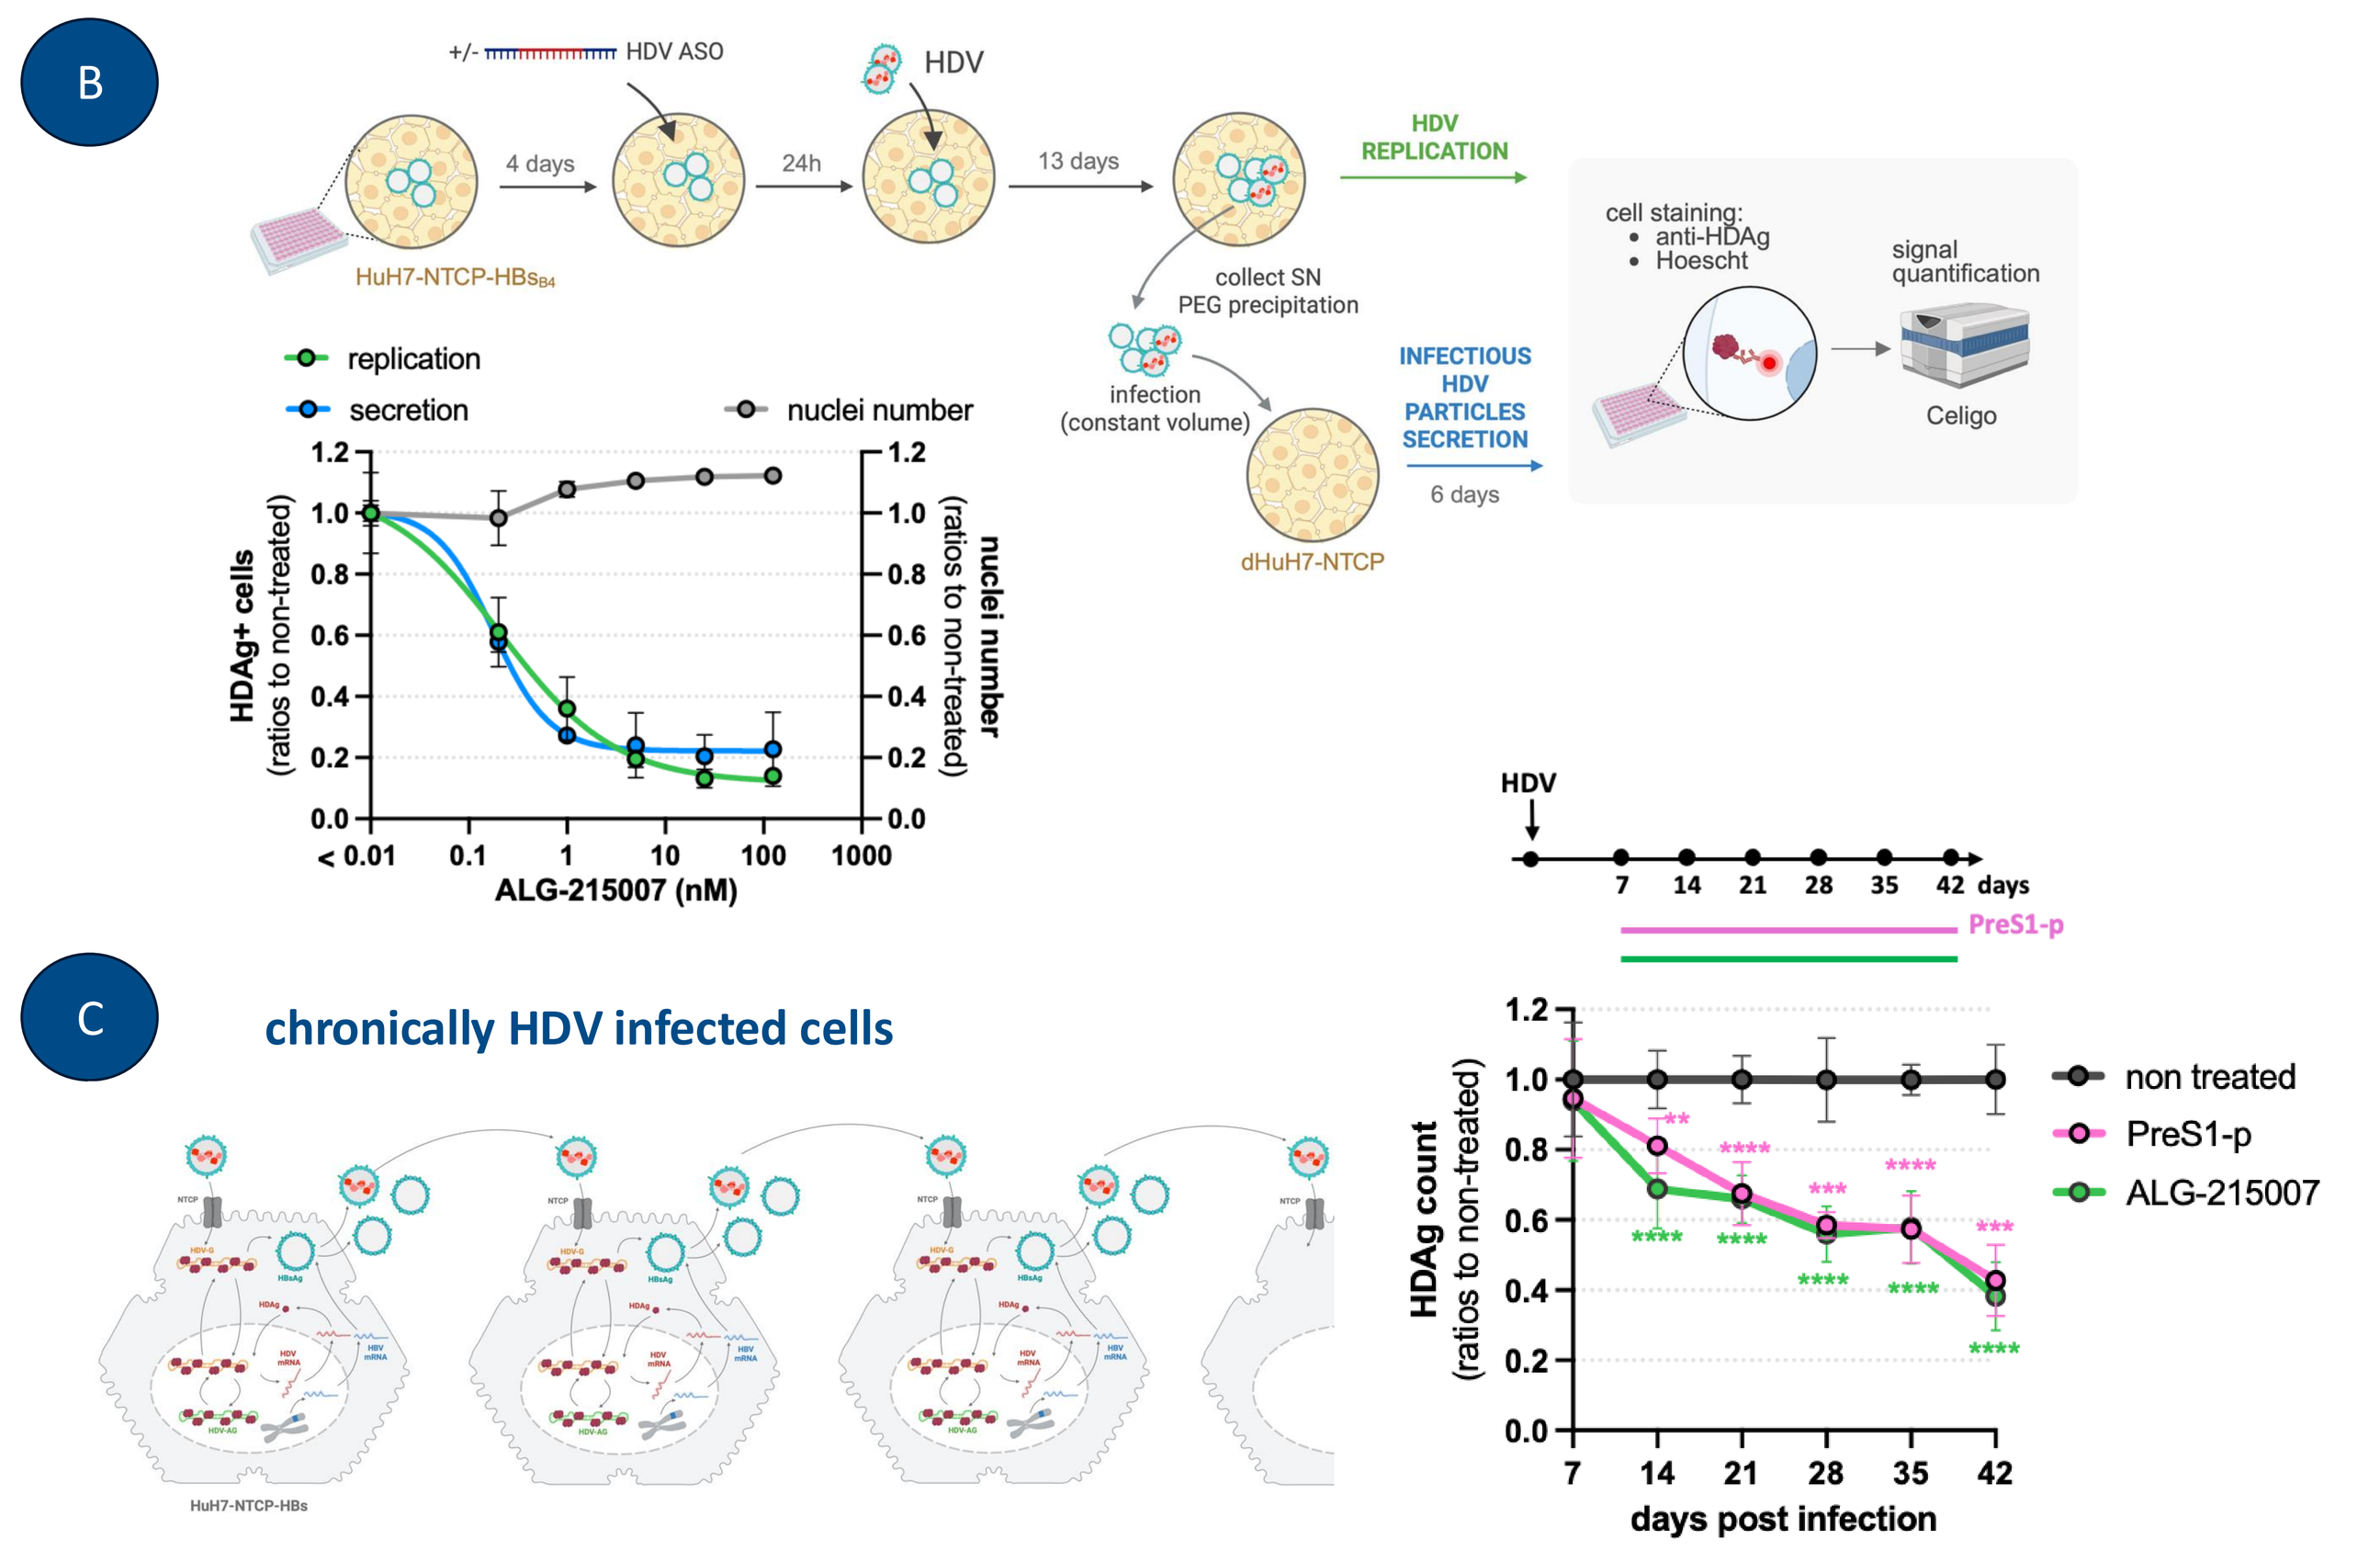
B
+/- HDV ASO
HDV
4 days
24h
13 days
HuH7-NTCP-HBs
HDV REPLICATION
collect SN
PEG precipitation
infection (constant volume)
INFECTIOUS HDV PARTICLES SECRETION
6 days
dHuH7-NTCP
cell staining:
anti-HDAg
Hoescht
signal quantification
Celigo
replication
secretion
nuclei number
HDAg+ cells (ratios to non-treated)
nuclei number (ratios to non-treated)
ALG-215007 (nM)
C
chronically HDV infected cells
HuH7-NTCP-HBs
HDV
PreS1-p
HDAg count (ratios to non-treated)
non treated
PreS1-p
ALG-215007
days post infection

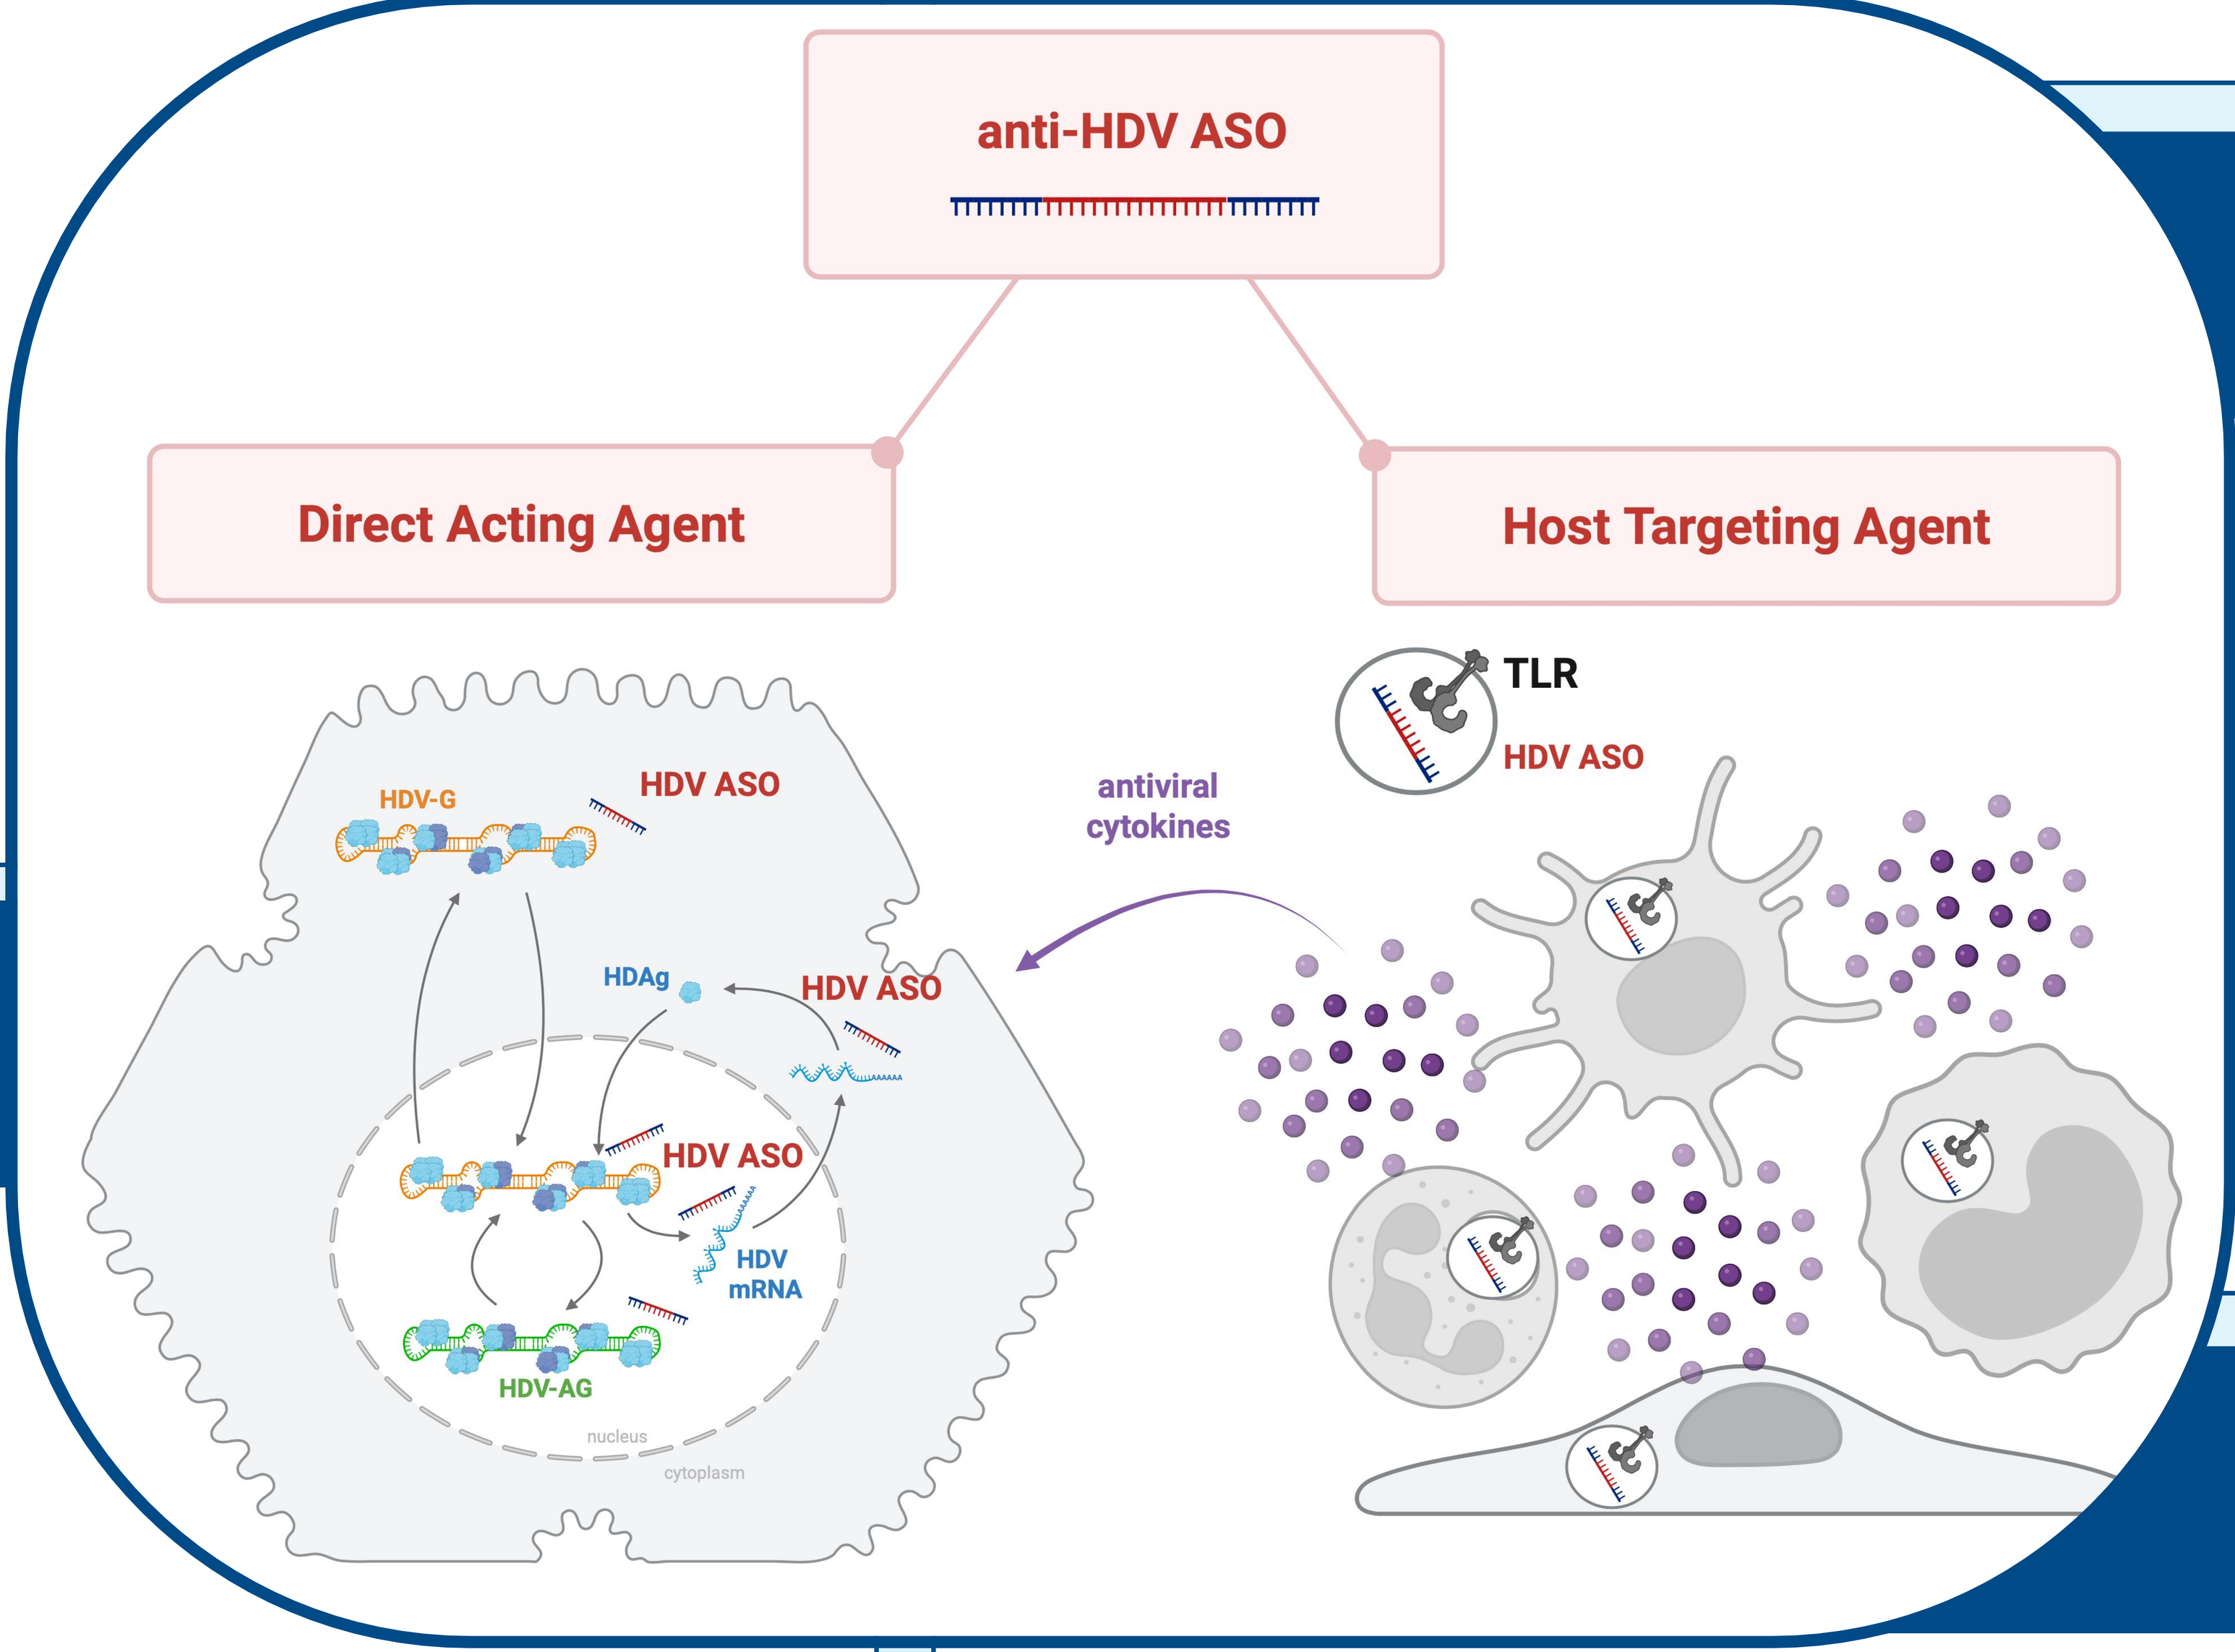
anti-HDV ASO
Direct Acting Agent
Host Targeting Agent
HDV-G
HDV ASO
HDAg
HDV mRNA
HDV-AG
nucleus
cytoplasm
TLR
antiviral cytokines

3. ALG-215007 targeting of HDV-AG

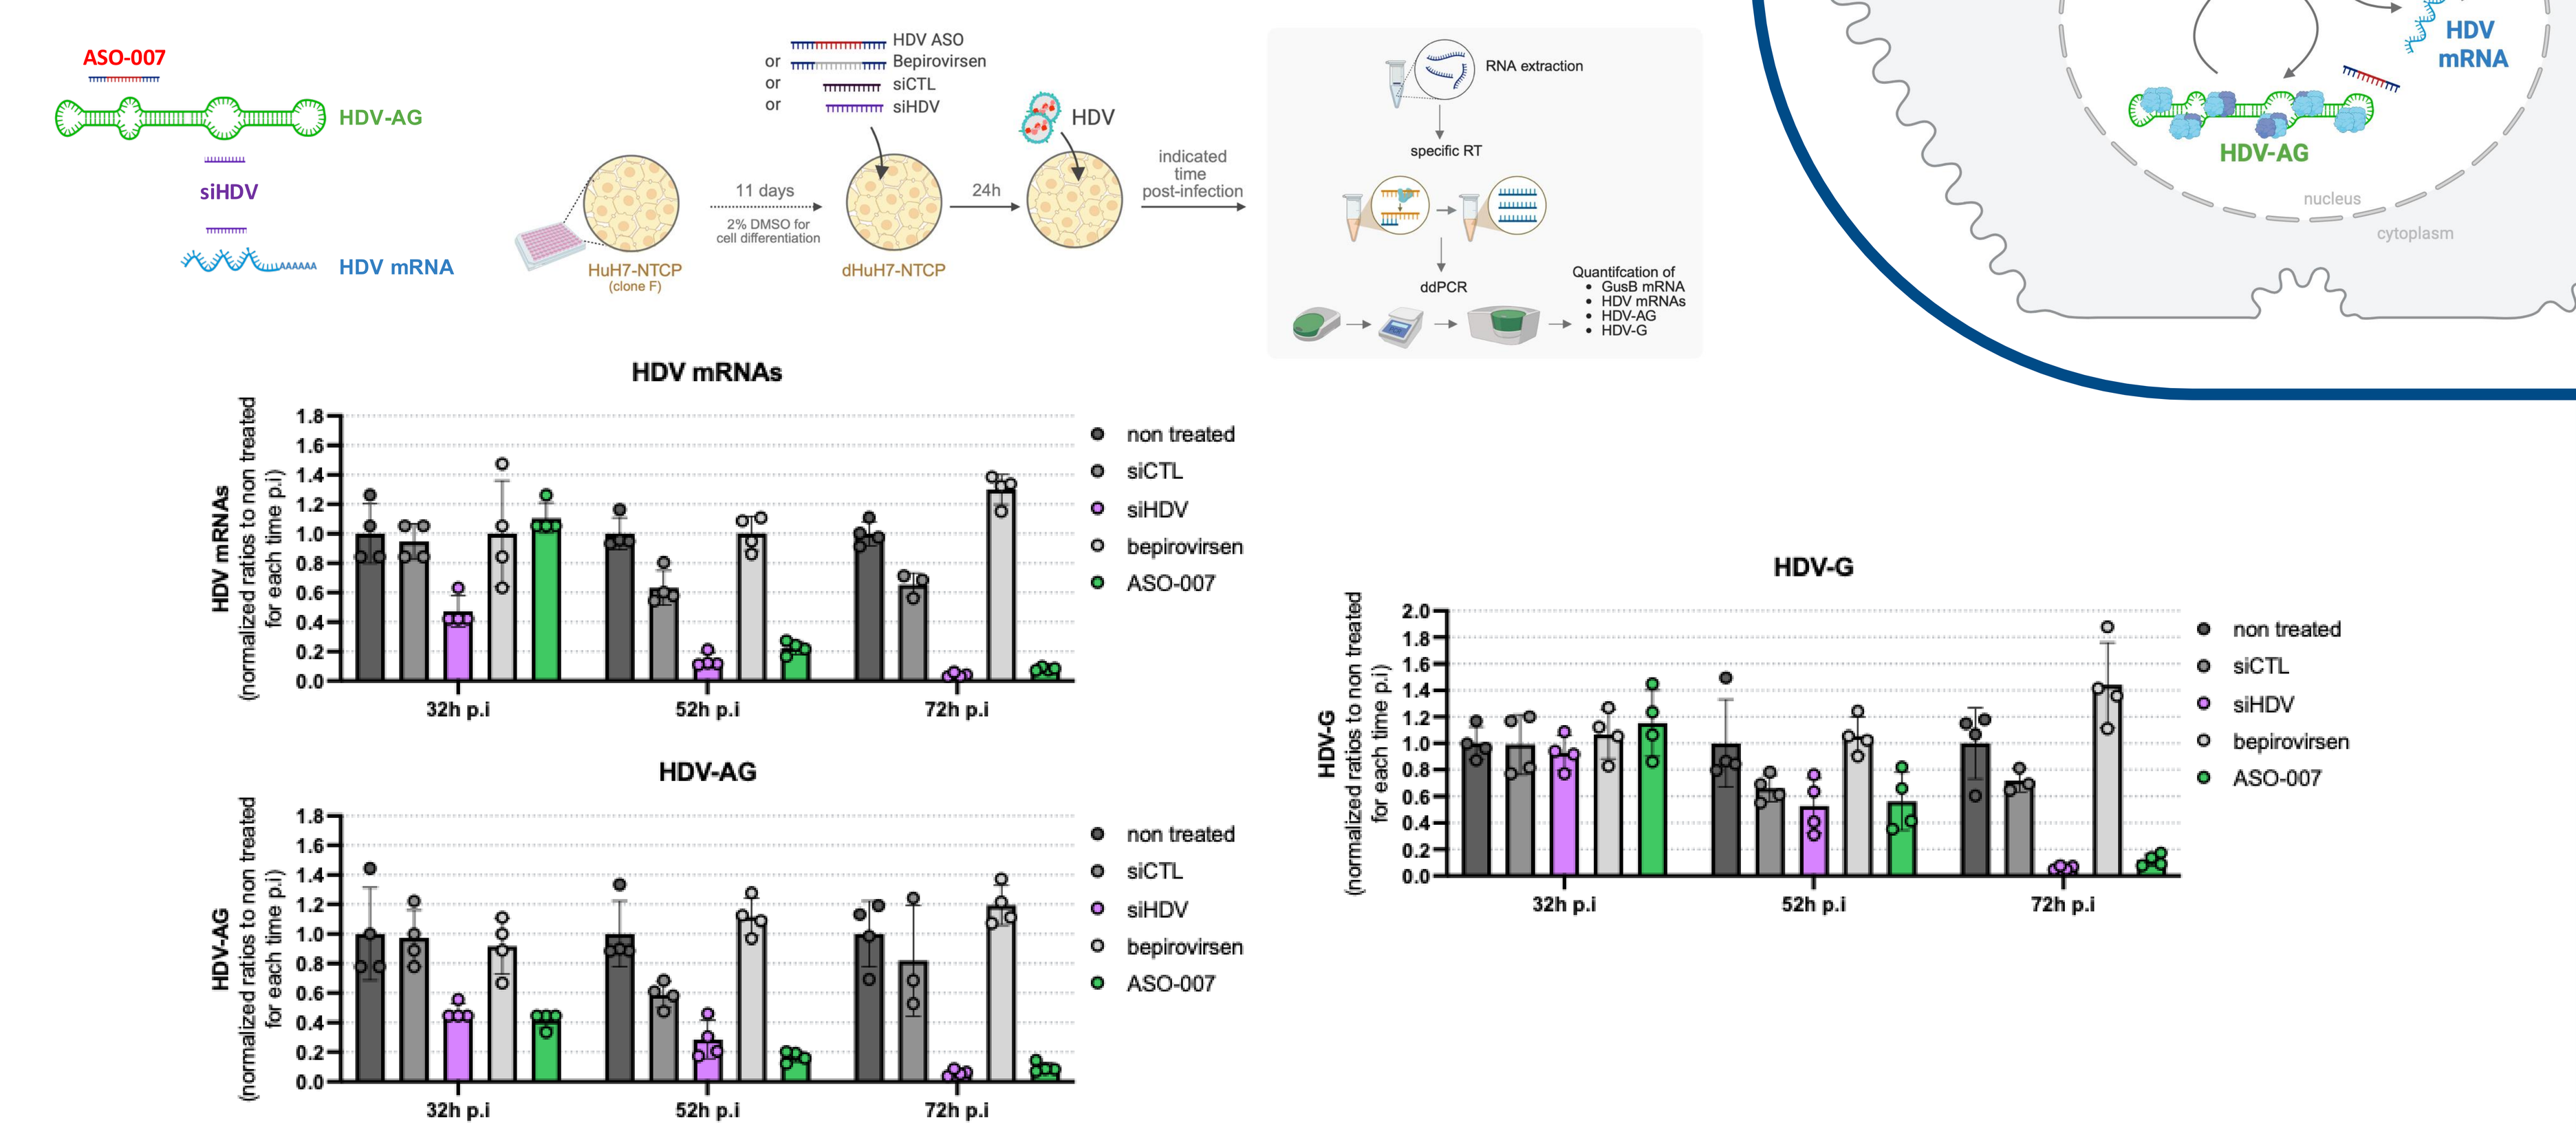
ASO-007
HDV-AG
siHDV
HDV mRNA
HDV ASO
or Bepirovirsen
or siCTL
or siHDV
HDV
11 days
2% DMSO for cell differentiation
24h
indicated time post-infection
HuH7-NTCP (clone F)
dHuH7-NTCP
RNA extraction
specific RT
ddPCR
Quantifcation of
GusB mRNA
HDV mRNAs
HDV-AG
HDV-G
HDV mRNAs
HDV-AG
HDV-G
non treated
siCTL
siHDV
bepirovirsen
ASO-007
32h p.i
52h p.i
72h p.i

4. Slight induction of hTLR8 by ALG-215007

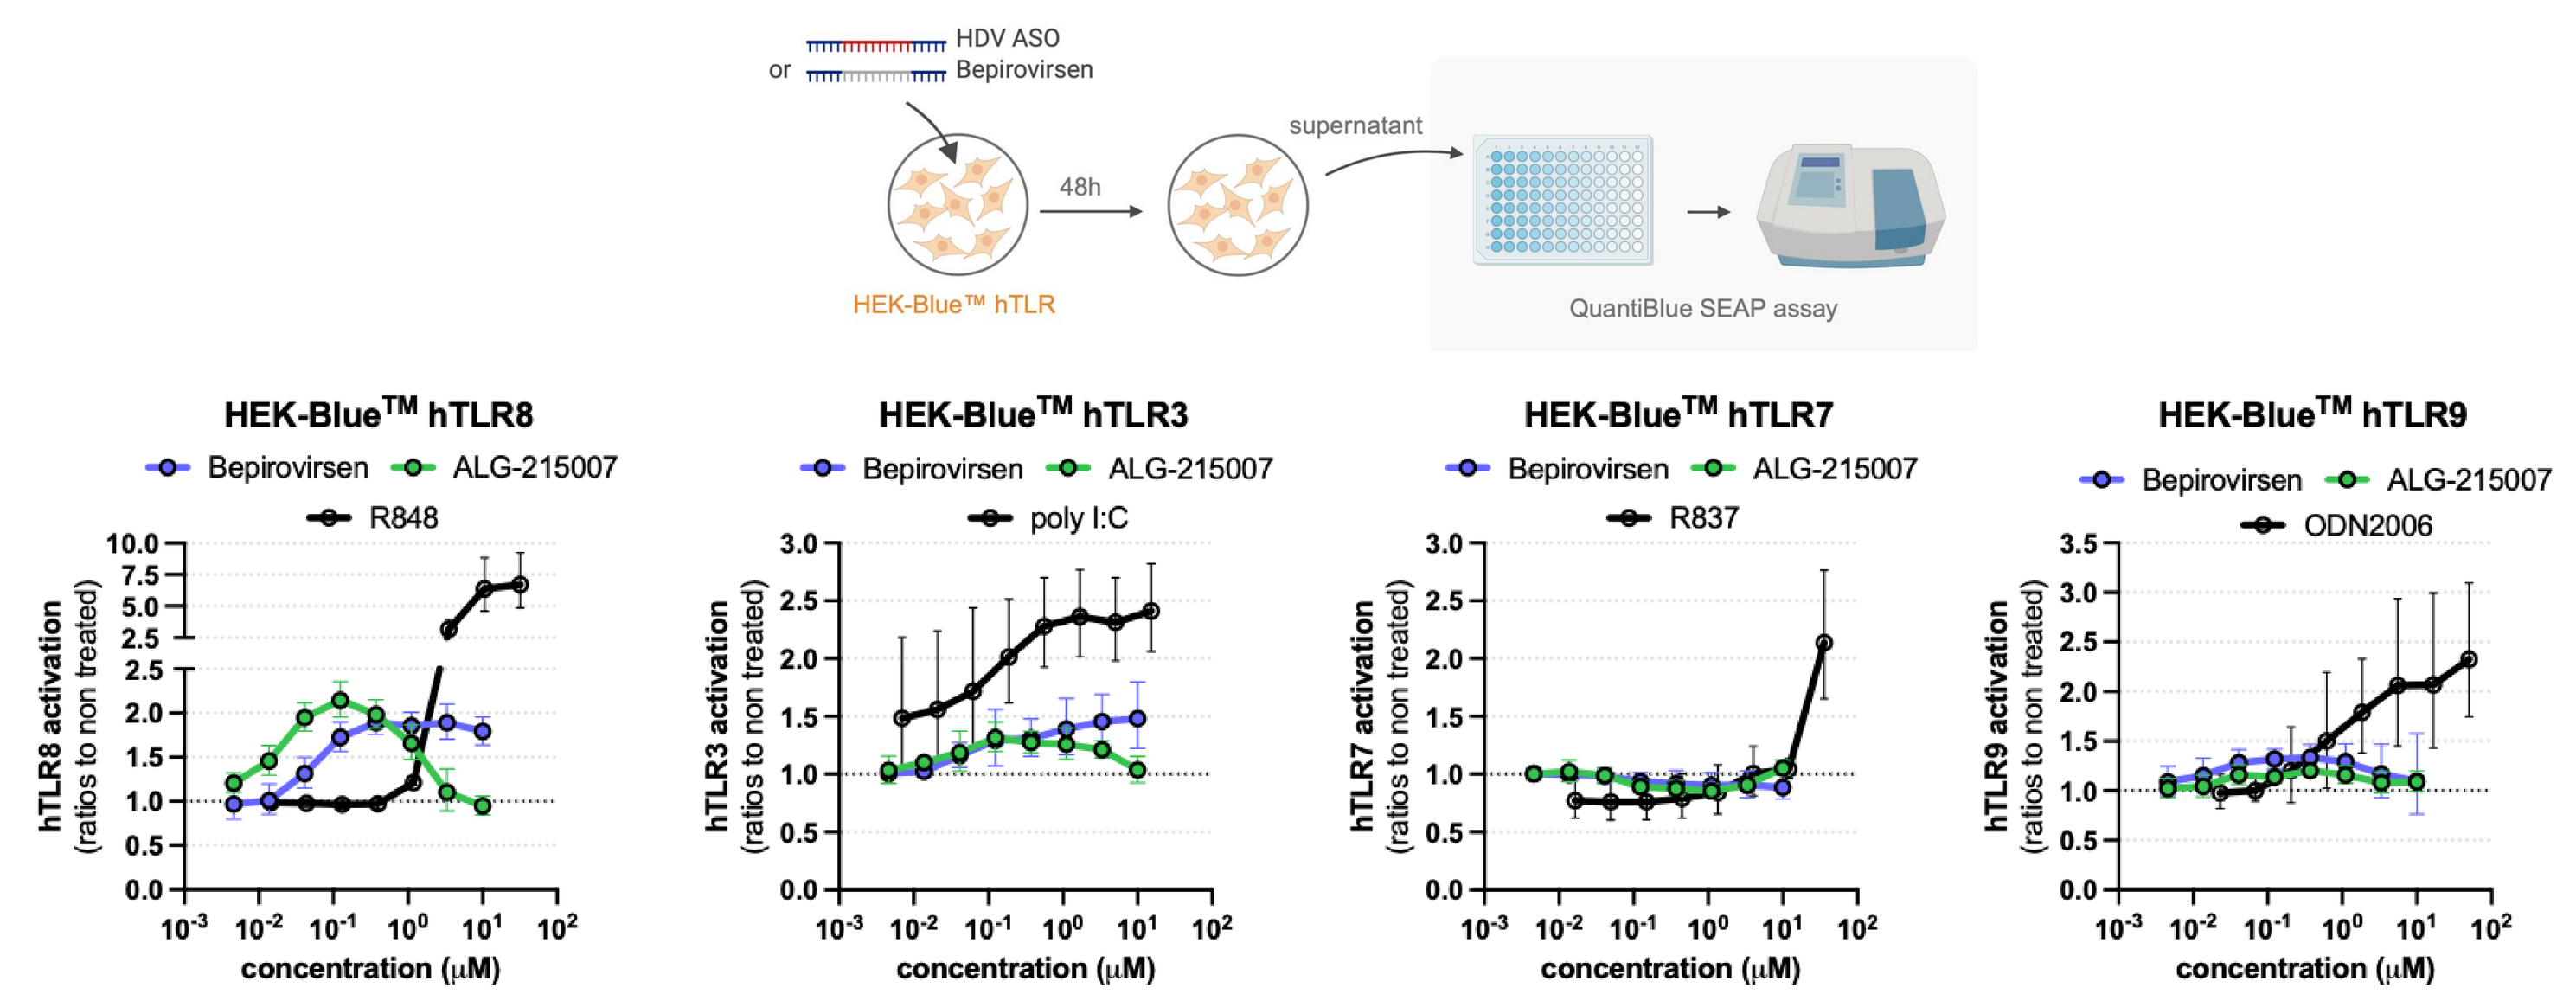
HDV ASO
or Bepirovirsen
48h
supernatant
HEK-Blue™ hTLR
QuantiBlue SEAP assay
HEK-Blue™ hTLR8
HEK-Blue™ hTLR3
HEK-Blue™ hTLR7
HEK-Blue™ hTLR9
Bepirovirsen
ALG-215007
R848
poly I:C
R837
ODN2006
concentration (μM)

5. Design for *in vivo* evaluation of ALG-215007

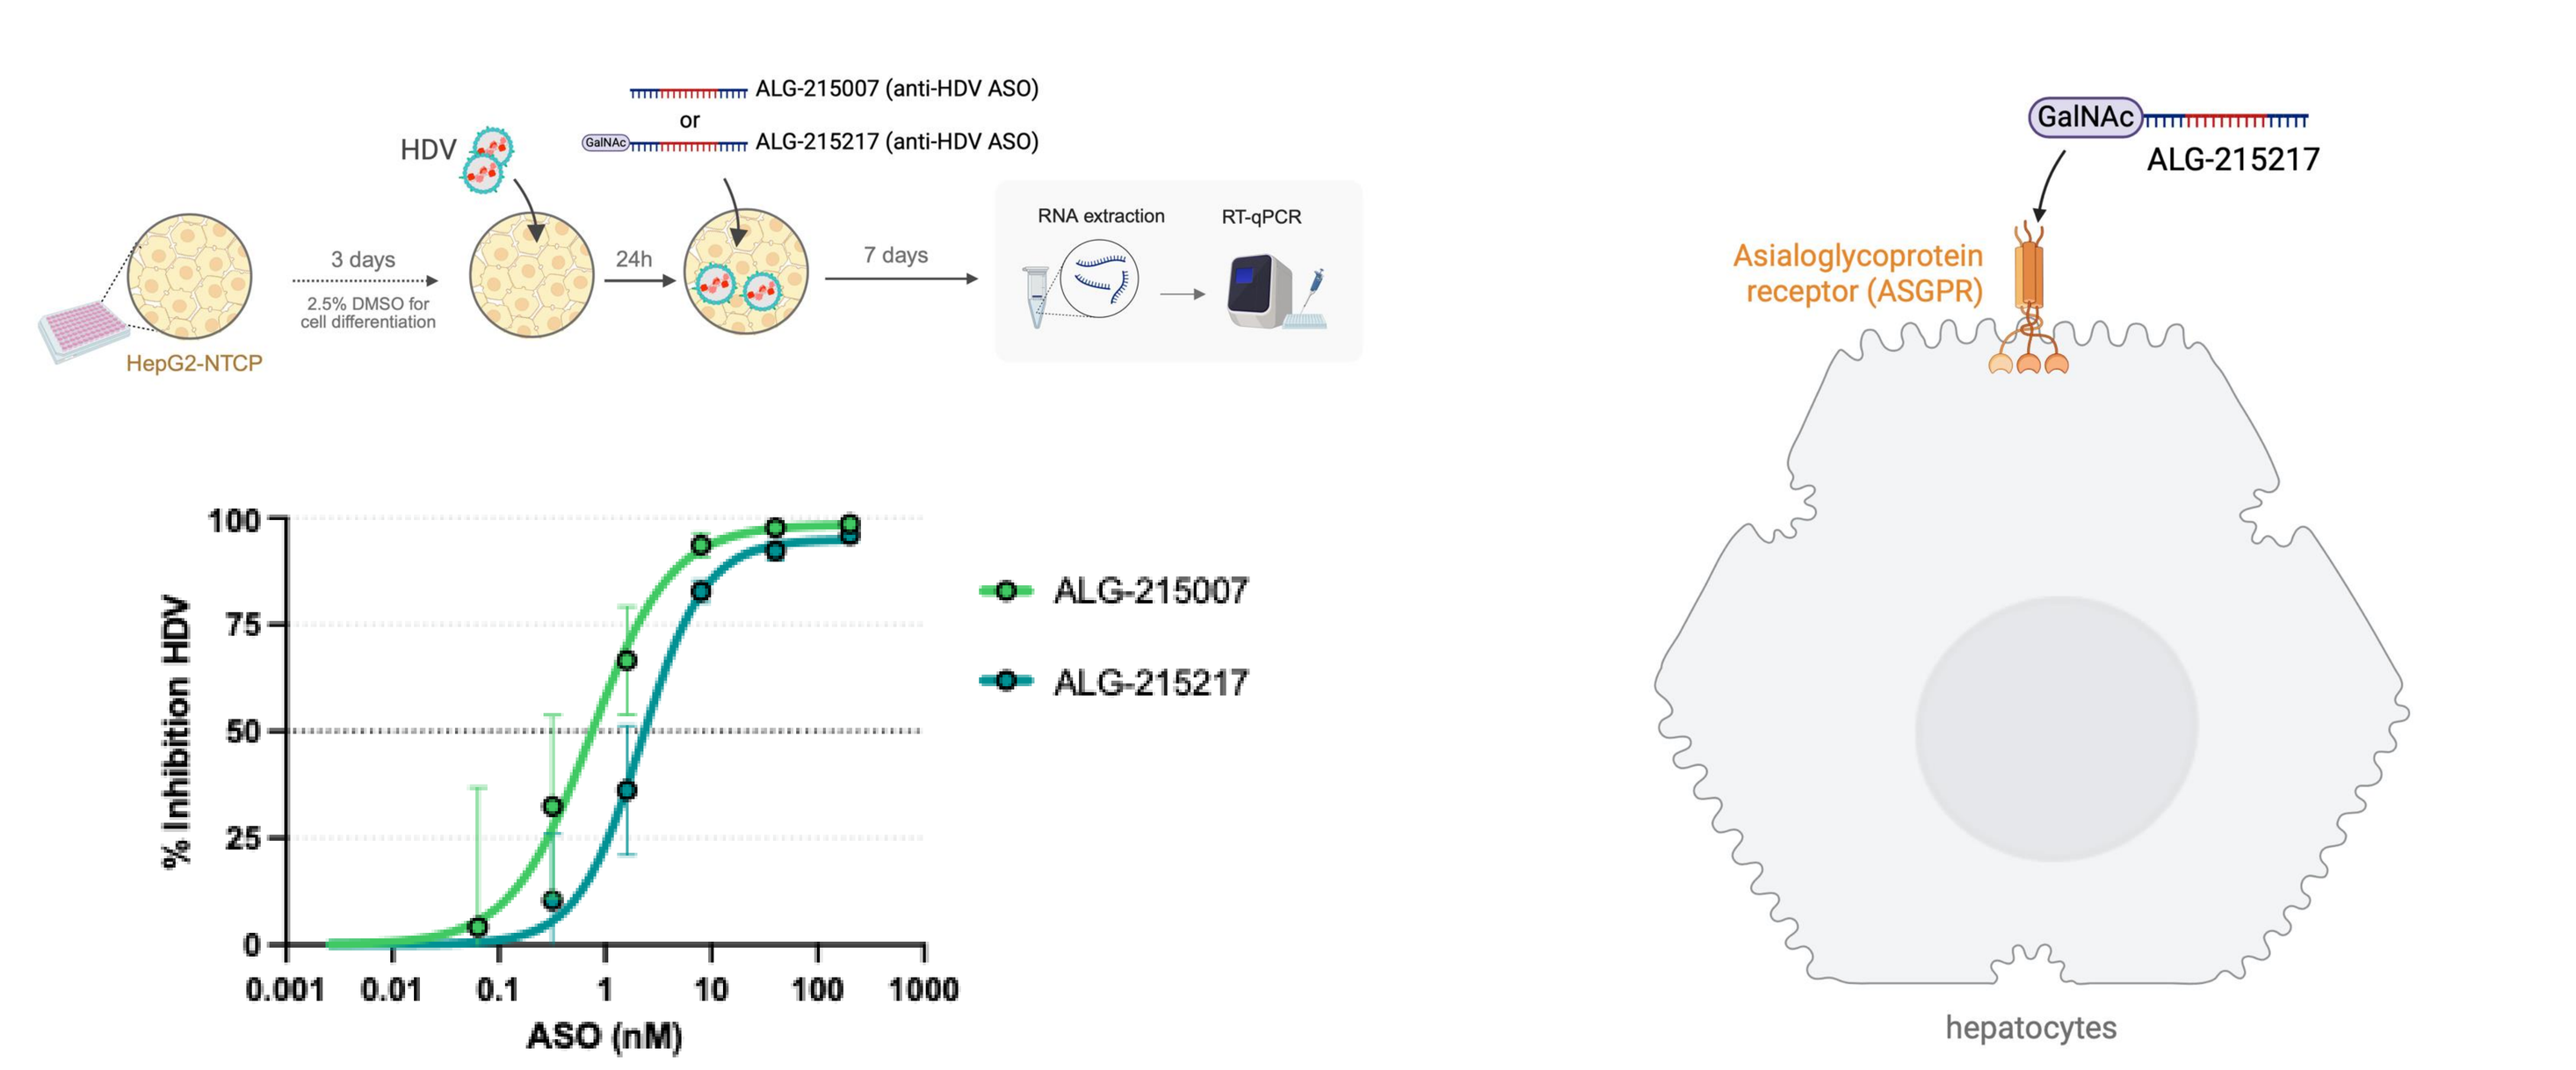
ALG-215007 (anti-HDV ASO)
or
ALG-215217 (anti-HDV ASO)
HDV
HepG2-NTCP
3 days
2.5% DMSO for cell differentiation
24h
7 days
RNA extraction
RT-qPCR
% Inhibition HDV
ASO (nM)
ALG-215007
ALG-215217
GalNAc
ALG-215217
Asialoglycoprotein receptor (ASGPR)
hepatocytes

Experiment currently ongoing

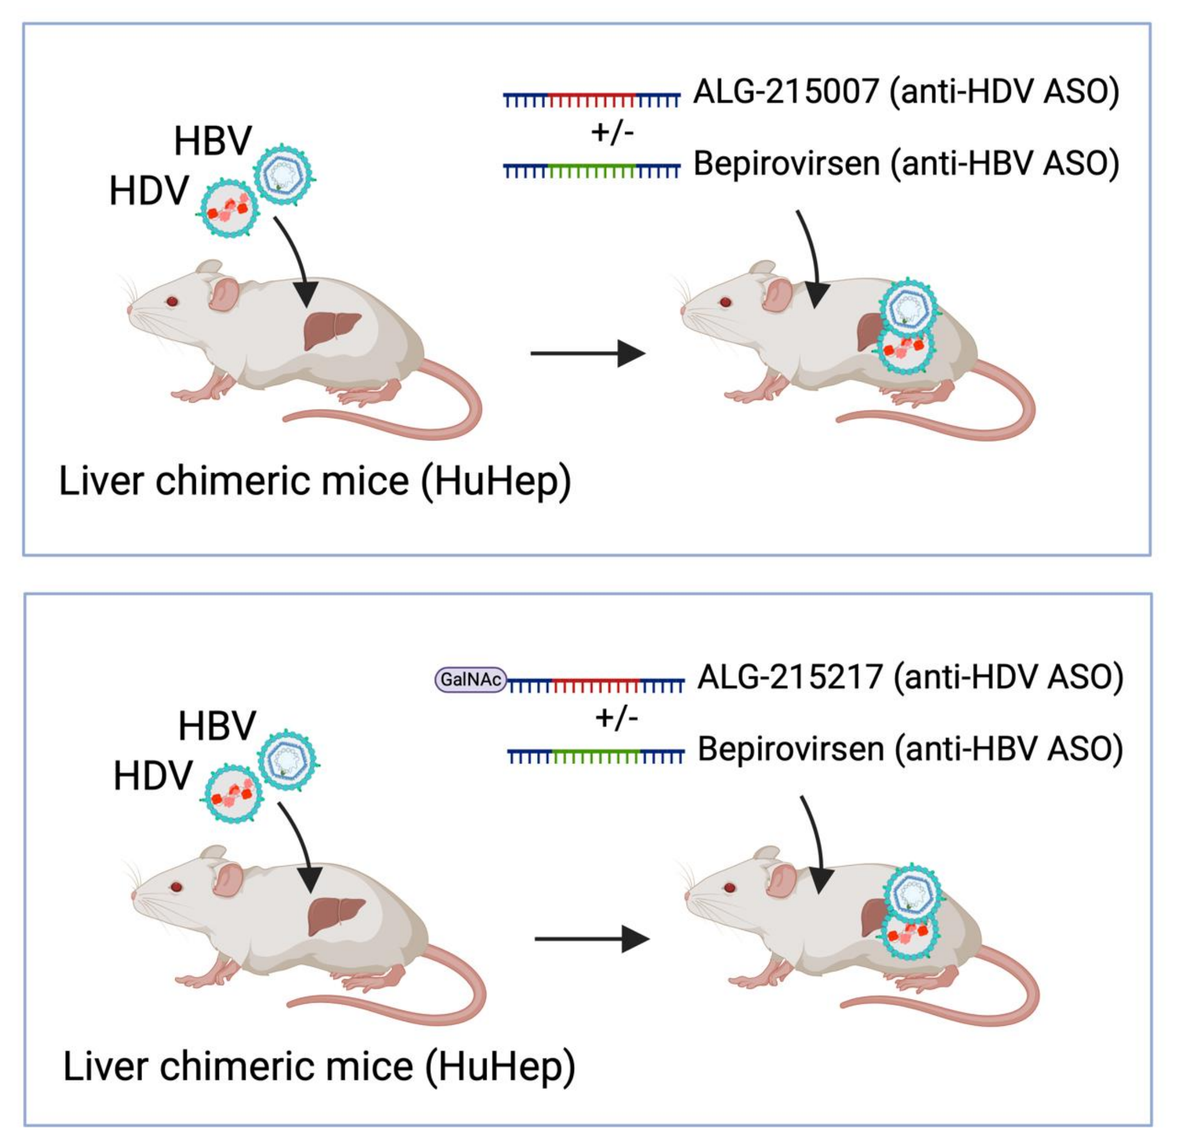
HBV
HDV
ALG-215007 (anti-HDV ASO)
+/-
Bepirovirsen (anti-HBV ASO)
Liver chimeric mice (HuHep)
ALG-215217 (anti-HDV ASO)
+/-
Bepirovirsen (anti-HBV ASO)
Liver chimeric mice (HuHep)

Acknowledgments

Figures created with www.biorender.com