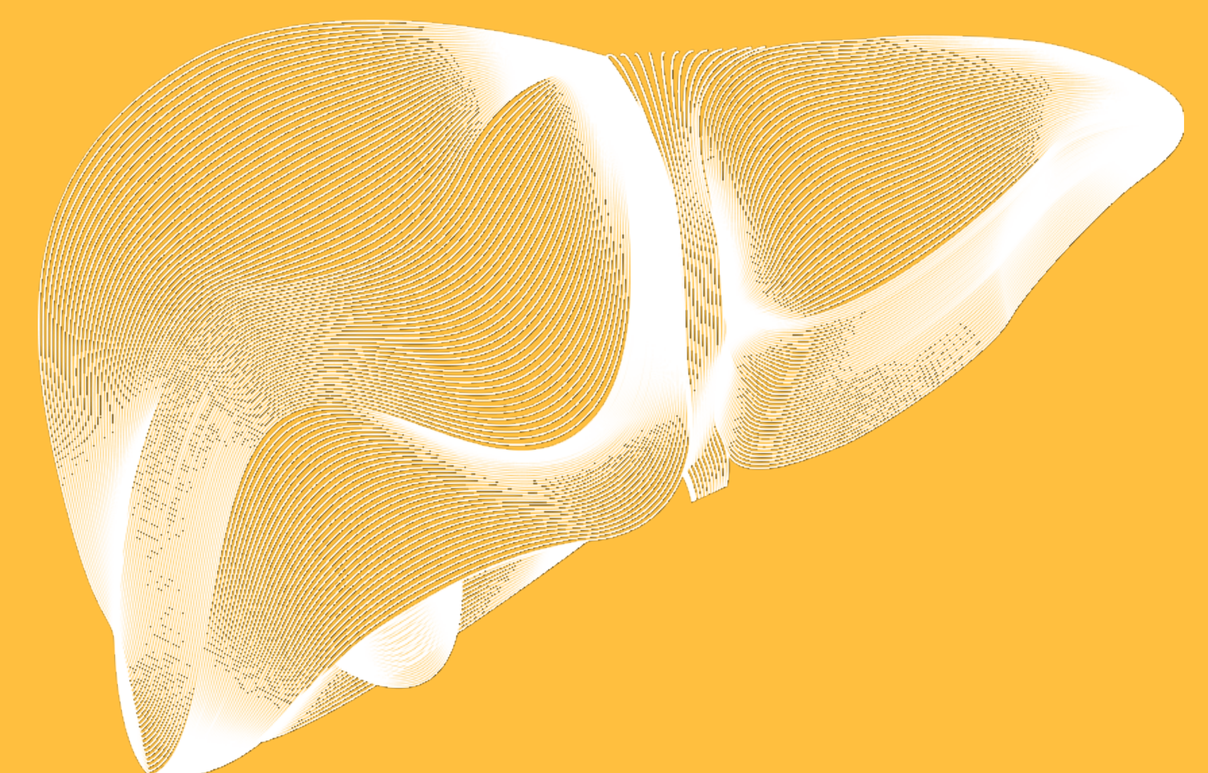

Extended Treatment of HBeAg+ CHB Subjects with the Capsid Assembly Modulator ALG-000184 with or without Entecavir is Associated with Reductions in Viral Markers and Favorable Anti-HBeAg trends

Man-Fung Yuen[1], Edward Gane[2], Kosh Agarwal[3], Junqi Niu[4], Hong Ren[5], Yanhua Ding[4], Xieer Liang[6], Xian Yu[5], Jia Xu[4], Alina Jucov[7], Alexei Haceatrean[7], Min Wu[8], Kha Le[8], Maida Maderazo[8], Lawrence M. Blatt[8], Sushmita Chanda[8], Tse-I Lin[8], Matthew McClure[8] and Jinlin Hou[6]

[1]Department of Medicine, School of Clinical Medicine, The University of Hong Kong, [2]Faculty of Medicine, University of Auckland, [3]King's College Hospital, Institute of Liver Studies, [4]The First Hospital of Jilin University, [5]The Second Affiliated Hospital of ChongQing Medical University, [6]Nanfang Hospital, Southern Medical University, [7]ARENSIA Exploratory Medicine, Republican Clinical Hospital and Nicolae Testemitanu State University of Medicine and Pharmacy, Moldova, [8]Aligos Therapeutics, Inc.

EASL CONGRESS 2024 Milan, Italy 5–8 June

WED-365

Contact information
Min Wu mwu@aligos.com

Introduction

- The sustained HBV DNA suppression (< lower limit of quantitation [LLOQ]) in chronic hepatitis B (CHB) patients with antiviral treatment has been associated with favorable disease outcomes[1].
- ALG-000184 is a prodrug of ALG-001075, a novel, pan-genotypic CAM-E (empty capsids) with high in vitro potency.
- The dual mechanism of action (MOA) of ALG-001075 has been demonstrated in vitro: 1) inhibition of pg-RNA encapsidation (1st MOA); 2) inhibition of HBV antigen production through blocking cccDNA establishment/replenishment at higher concentrations (2nd MOA)[2].
- In the ongoing Study ALG-000184-201, a favorable safety profile and multiple $\log_{10}$ reductions in HBV DNA/RNA and HBV antigens (HBs, HBe, HBcr) in HBeAg-positive (HBeAg+) CHB subjects received 300 mg ALG-000184 $\pm$ entecavir (ETV) $\leq$ 64 weeks were previously reported[3].

Aim

To evaluate the safety, pharmacokinetics (PK), and antiviral activity of long-term treatment with ALG-000184 in untreated CHB subjects

Method

ALG-000184-201 (NCT04536337) is a multipart Phase 1 study. Parts 1-3 and Part 4 Cohort 1 are complete, and data have been reported previously [4,5,6]. Part 4 Cohort 2 and B are ongoing, and the study designs are shown in Figure 1. Briefly, these 2 cohorts assess the safety, PK, & antiviral activity of an oral daily 300 mg dose of ALG-000184 with (Cohorts 2) or without (Cohort B) ETV in HBeAg+ (Cohorts 2 & B) or HBeAg-negative (HBeAg-) (Cohort B) CHB subjects. The planned dosing duration is 96 weeks. An active comparator ETV monotherapy x 12 weeks was included in Cohort 2. Here we update the data from the extended treatment of 300 mg ALG-000184 ± ETV for up to 72 weeks in untreated HBeAg+ CHB subjects in Part 4 Cohorts 2 (P4C2) and B (P4CB).

Figure 1 ALG-000184-201 Part 4 Cohort 2 and B Design

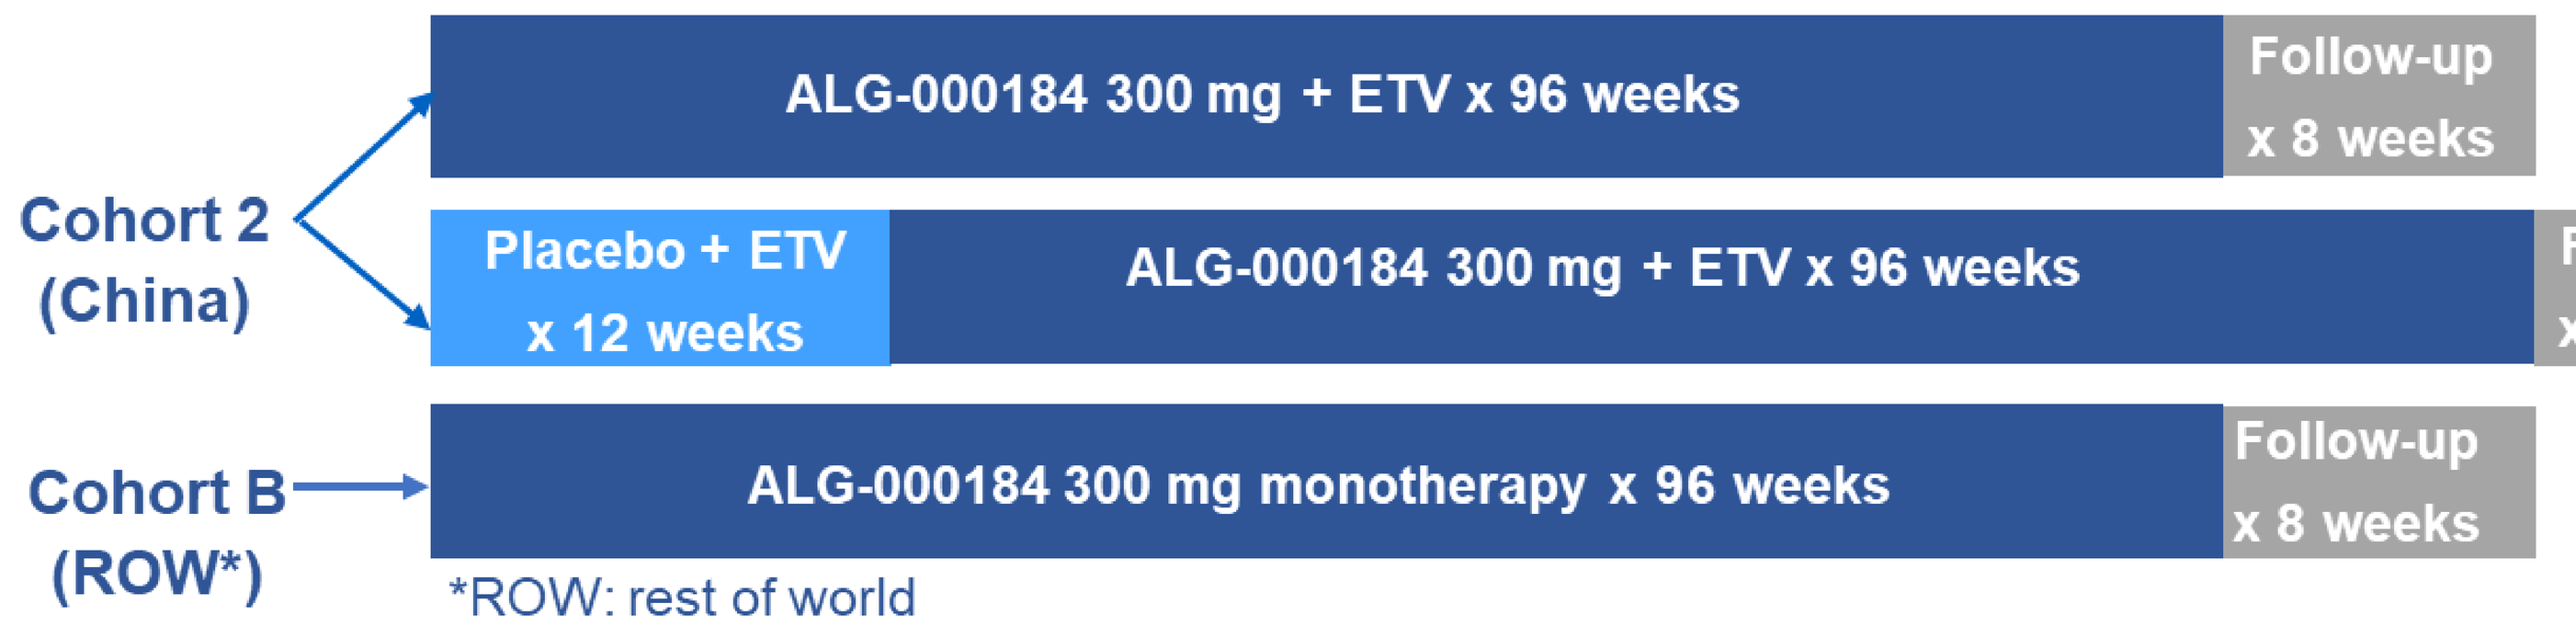
Cohort 2 (China)
ALG-000184 300 mg + ETV x 96 weeks
Follow-up x 8 weeks
Placebo + ETV x 12 weeks
ALG-000184 300 mg + ETV x 96 weeks
Follow-up x 8 weeks
Cohort B (ROW*)
ALG-000184 300 mg monotherapy x 96 weeks
Follow-up x 8 weeks

*ROW: rest of world

Throughout the study, safety assessments (adverse events [AEs], vital signs, electrocardiogram [ECG] and laboratories), PK, and viral markers were collected at regular intervals. The Study Review Committee (SRC) and ALT Flare Committee (AFC) had safety oversight and reviewed safety and PK data on a regular basis.

Virology markers were analyzed in two central laboratories: Sonic for Rest-of-World (ROW) sites and KingMED for China sites. Lower Limit of Quantitative (LLOQ) and Detection (LLOD) of HBV DNA are 10 IU/mL and ≤ 4.92 IU/mL, respectively. LLOQs of HBV RNA, HBsAg, HBeAg and HBcrAg are 10 (ROW) or 200 (China) copies/mL, 0.05 IU/mL, 0.01 (ROW) or 0.22 (China) PEI U/mL and 3 $\log_{10}$ U/mL, respectively. P4C2 & P4CB virology summaries only include data at time points where subjects were confirmed (by PK) being compliant on taking 300 mg ALG-000184 + ETV.

Result

Baseline Characteristics

- A total of 25 HBeAg+ CHB subjects were enrolled and received 300 mg ALG-000184 + ETV (n=15, P4C2) or 300 mg ALG-000184 monotherapy (n=10, P4CB).
- Baseline characteristics were typical for untreated HBeAg+ population and comparable between P4C2 and P4CB (Table 2) .

Table 2: Baseline Characteristics

Characteristic	P4C2 (n=15*) 300mg ALG-000184 + ETV	P4CB (n=10) 300mg ALG-000184
Age, years, mean (SEM)	31.4 (3.3)	36.8 (2.9)
Male, n(%)	8 (53)	7 (70)
Asian, n(%)	3 (100)	9 (90)
BMI, kg/m^2, mean (SEM)	22.2 (0.8)	22.4 (0.8)
HBV Genotype, n(%)	B: 5 (33); C: 10 (67)	B: 5 (50); C: 4 (40); D: 1 (10)
HBV DNA, $\log_{10}$ IU/mL, mean (SEM)	8.1 (0.2)	8.0 (0.2)
HBV RNA, $\log_{10}$ copies/mL, mean (SEM)	6.7 (0.3)	5.3 (0.4)
HBsAg, $\log_{10}$ IU/mL, mean (SEM)	4.4 (0.2)	4.3 (0.1)
HBeAg, $\log_{10}$ PEI U/mL, mean (SEM)	2.4 (0.1)	2.6 (0.3)
HBcrAg, $\log_{10}$ U/mL, mean (SEM)	8.3 (0.3)	8.3 (0.2)

SEM: standard error of mean. *Three subjects prematurely discontinued due to non-safety related personal reasons (n=2) and non-compliance of dosing (n=1)

Safety

- 300 mg ALG-000184 ± ETV for up to 72 weeks was generally well tolerated

	300mg ALG-000184 + ETV	300mg ALG-000184
	N=15	N=10
Serious Adverse Event (SAE)	0	0
Treatment Emergent Adverse Event (TEAE) leading to study drug discontinuation	0	0
Grade ≥3 TEAE	5*,#	3#
Concerning laboratory, ECG, vital sign, or physical examination findings	0	0

* 3 subjects experienced Grade ≥ 3 TEAEs of asymptomatic laboratory abnormalities including eGRF decrease, uric acid increase and neutrophil count decrease. All had normalized or returned to baseline in setting of continued study dosing.

\# 3 subjects in Part 4 Cohort 2 and 3 subjects in Part 4 Cohort B experienced Grade ≥ 3 ALT ↑ with or without associated AST ↑. All were asymptomatic and none were associated with hepatic synthetic dysfunction or considered concerning to the study's ALT Flare Monitoring Committee. All resolved (n=5) or improved (n=1) in setting of continued study dosing.

Antiviral Activity: *HBV DNA and HBV RNA*

- 300 mg ALG-000184 ± ETV up to 72 weeks of treatment led to substantial HBV DNA reduction with a maximum mean reduction of 7.0 $\log_{10}$ IU/mL in HBeAg+ subjects.
- By Week 48, 6 of 10 subjects (60%) receiving 300 mg ALG-000184 monotherapy and 4 of 12 subjects (33%) receiving 300 mg ALG-000184 + ETV achieved HBV DNA suppression (< LLOQ 10 IU/mL). (Figure 3).
- By Week 72, 9 of 10 subjects (90%) received 300 mg ALG-000184 monotherapy and 6 of 12 subjects (50%) received 300 mg ALG-000184 + ETV achieved HBV DNA suppression (< LLOQ 10 IU/mL, Figure 2).
- The time to HBV DNA < LLOQ was associated with baseline HBV DNA level. (Figure 3)
- 100% (n=22) HBeAg+ subjects receiving 300 mg ALG-000184 ± ETV x ≥ 12 weeks achieved RNA suppression (< LLOQ, 10 or 200 copies/mL) by Week 40. The mean maximum reduction of HBV RNA was 4.7 $\log_{10}$ copies/mL.
- No viral breakthrough was observed in any subject (Figure 2).

Figure 2: Individual HBV DNA Level Over Time

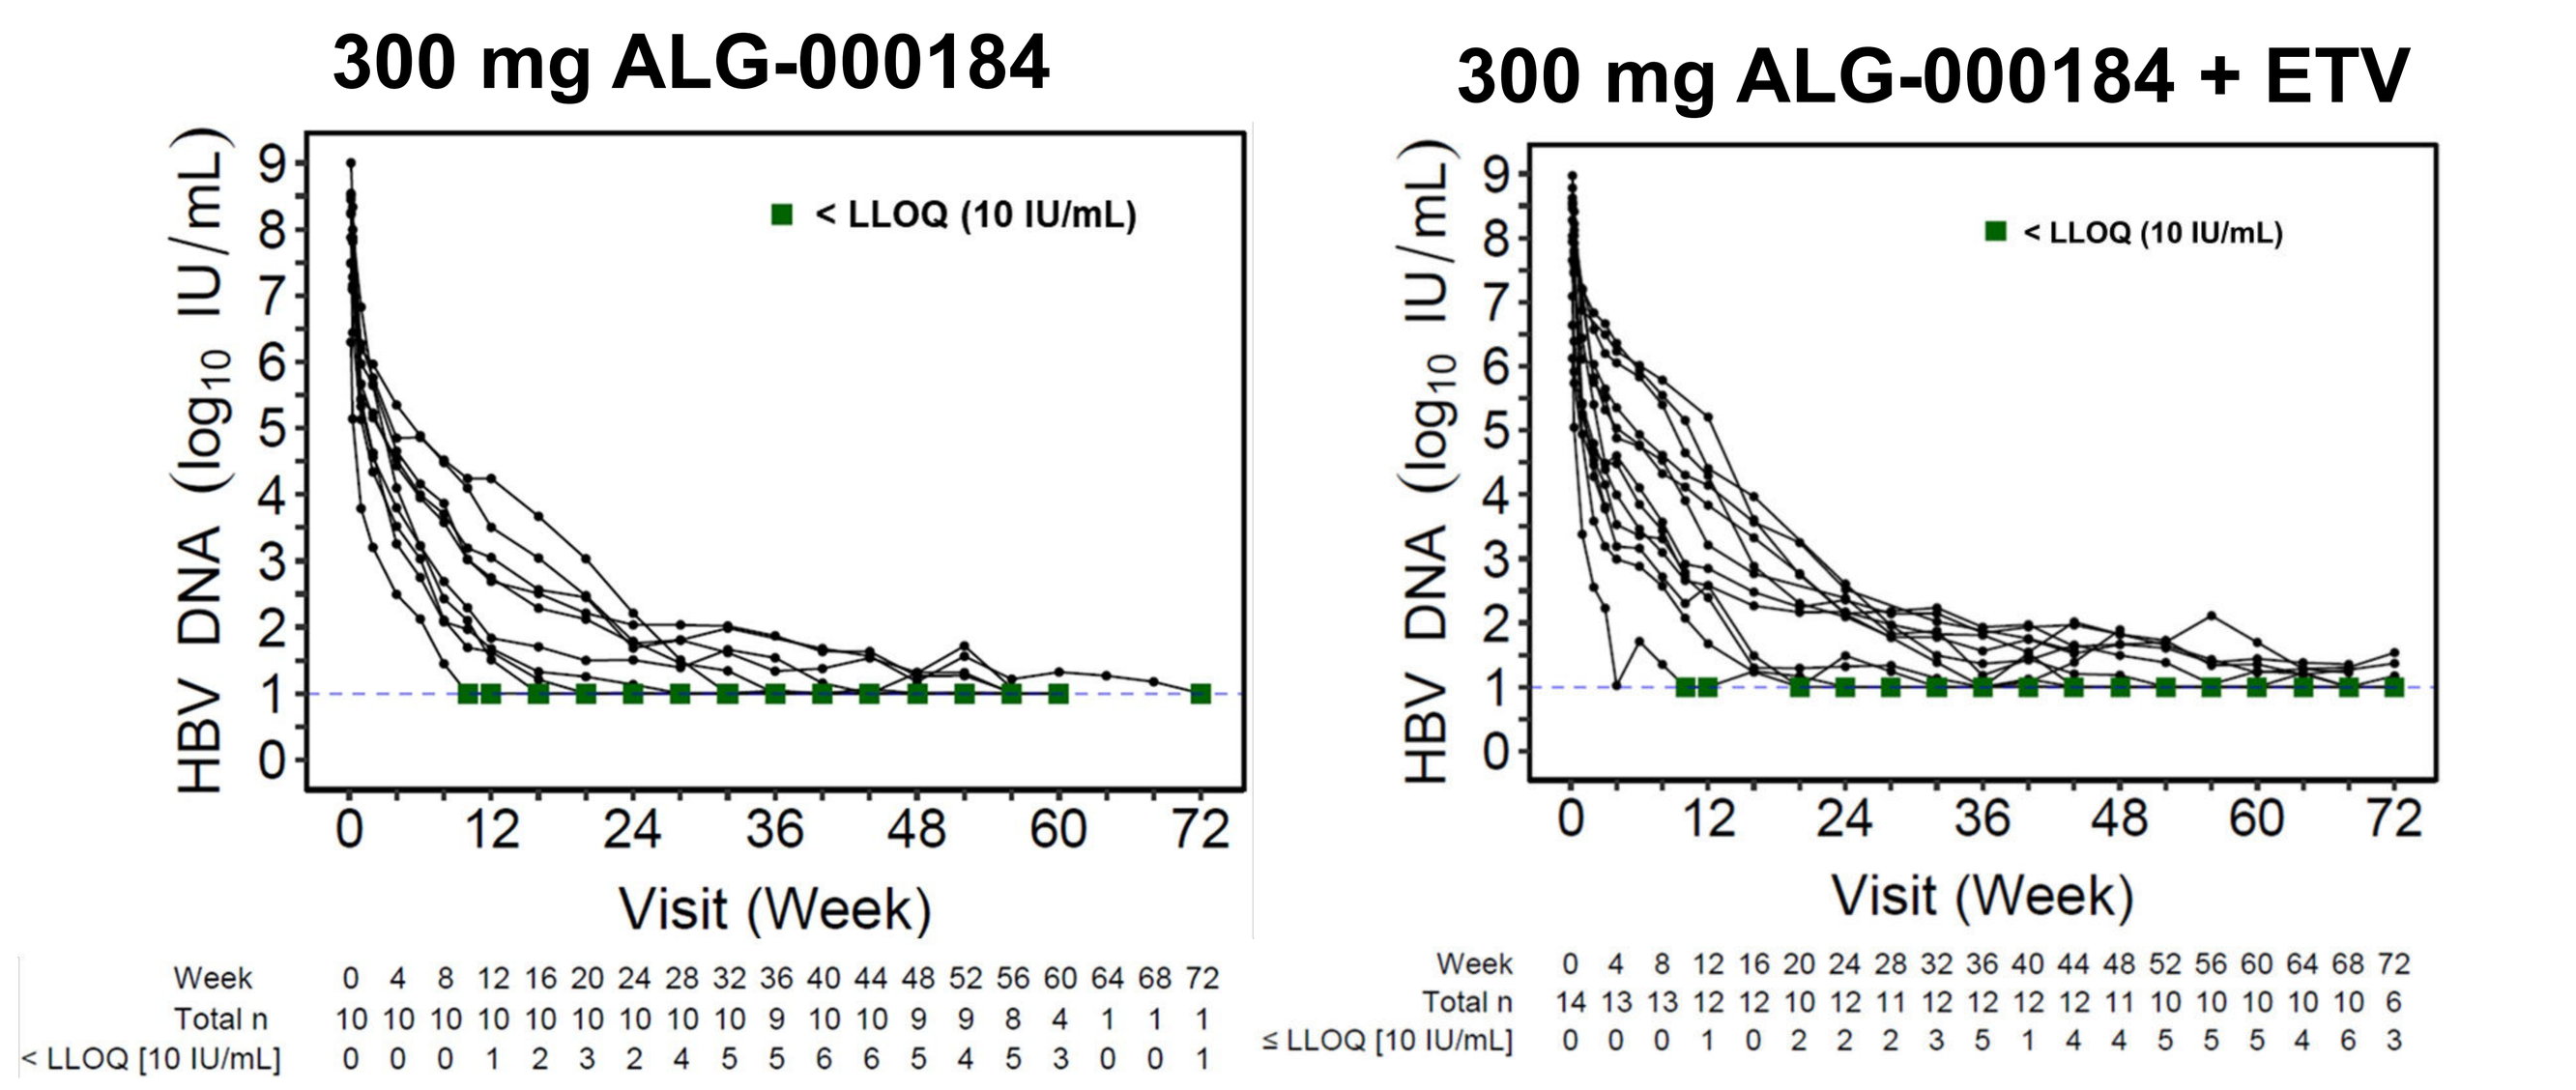
300 mg ALG-000184
300 mg ALG-000184 + ETV
< LLOQ (10 IU/mL)
HBV DNA (log10 IU/mL)
Visit (Week)
Week 0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72
Total n 10 10 10 10 10 10 10 10 10 9 10 10 9 9 8 4 1 1 1
< LLOQ [10 IU/mL] 0 0 0 1 2 3 2 4 5 5 6 6 5 4 5 3 0 0 1
Week 0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72
Total n 14 13 13 12 12 10 12 11 12 12 12 12 11 10 10 10 10 10 6
≤ LLOQ [10 IU/mL] 0 0 0 1 0 2 2 2 3 5 1 4 4 5 5 5 4 6 3

Figure 3: Individual time to HBV DNA < LLOQ 10 IU/mL

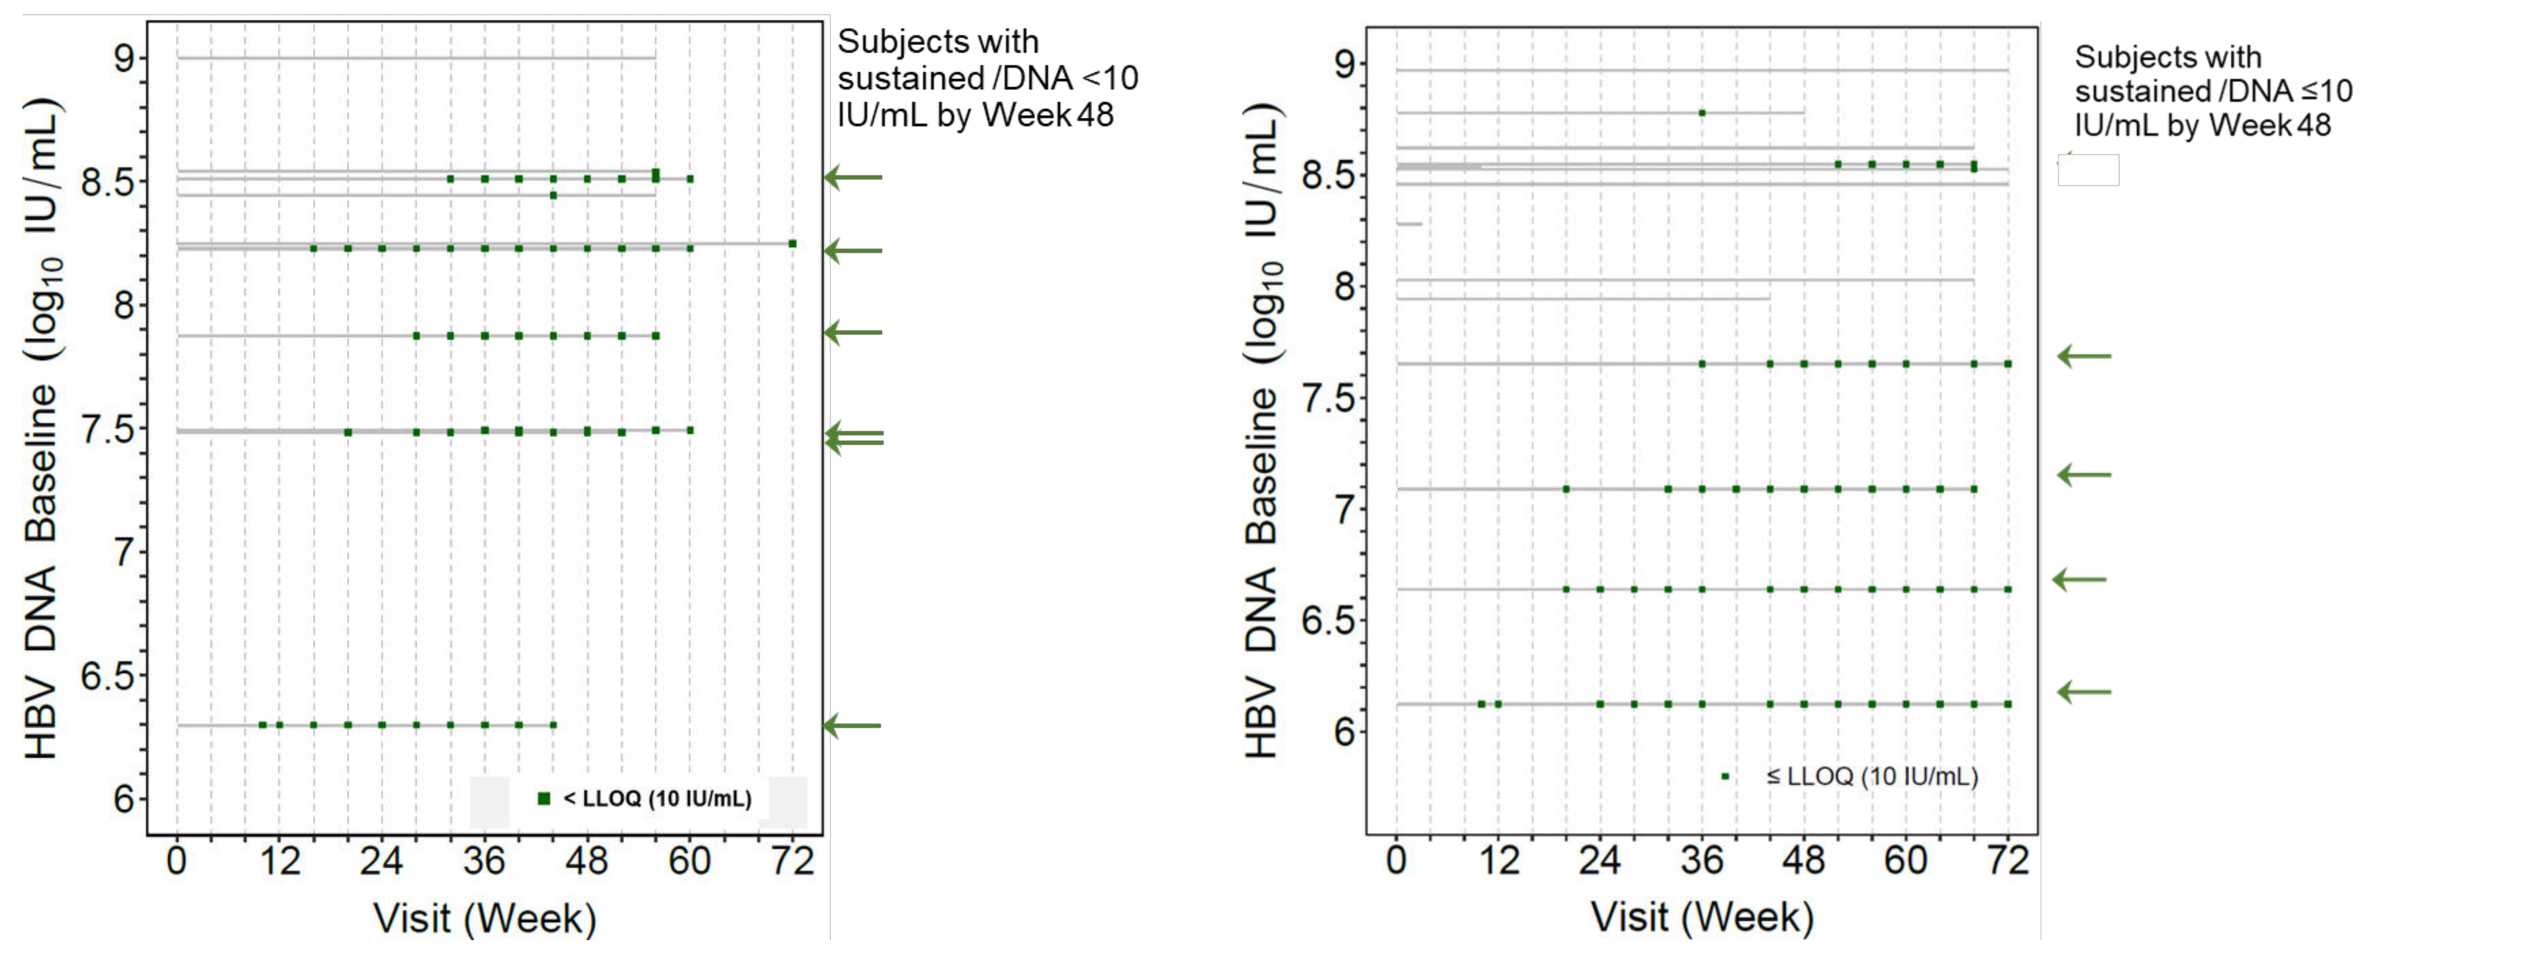
300 mg ALG-000184
300 mg ALG-000184 + ETV
Subjects with sustained /DNA <10 IU/mL by Week 48
Subjects with sustained /DNA ≤10 IU/mL by Week 48
HBV DNA Baseline (log10 IU/mL)
< LLOQ (10 IU/mL)
≤ LLOQ (10 IU/mL)
Visit (Week)

Antiviral Activity: *HBsAg, HBeAg, HBcrAg*

- Multi-$\log_{10}$ reductions in HBsAg, HBeAg and HBcrAg were observed in HBeAg+ subjects receiving 300 mg ALG-000184 ± ETV x ≤72 weeks (Figure 4)
- The mean maximum declines in HBsAg, HBeAg and HBcrAg were 1.0 $\log_{10}$ IU/mL, 1.9 PEI $\log_{10}$ U/mL and 2.1 $\log_{10}$ U/mL by 72 weeks of treatment, respectively.
- Anti-HBe antibody (HBeAb) level showed positive trend with decline of HBeAg (Figure 5) despite no subject experiencing HBeAg loss and/or anti-HBeAg positivity.

Figure 4: Mean $\log_{10}$ Reduction of HBsAg, HBeAg and HBcrAg from Baseline

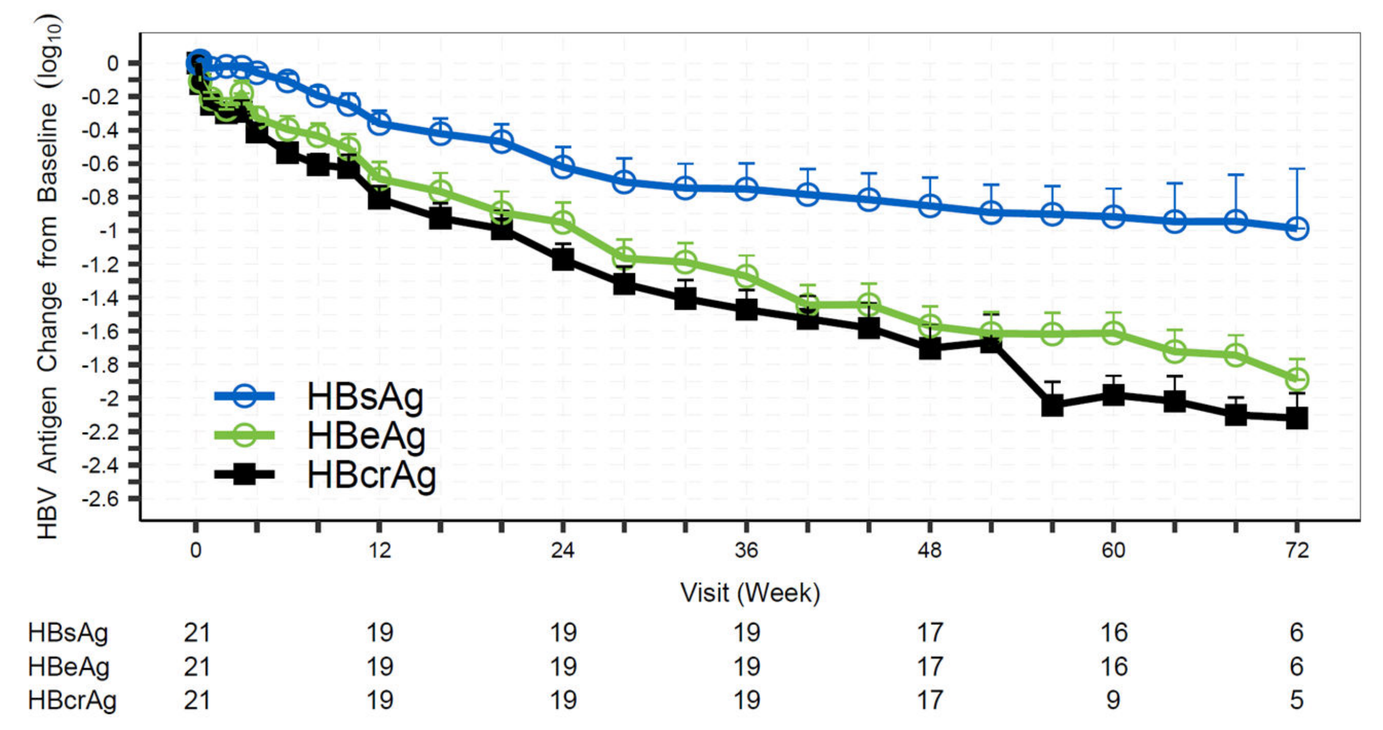
HBV Antigen Change from Baseline (log10)
HBsAg
HBeAg
HBcrAg
Visit (Week)
HBsAg 21 19 19 19 17 16 6
HBeAg 21 19 19 19 17 16 6
HBcrAg 21 19 19 19 17 9 5

Figure 5: Mean HBeAg and Anti-HBe Antibody Level Over time

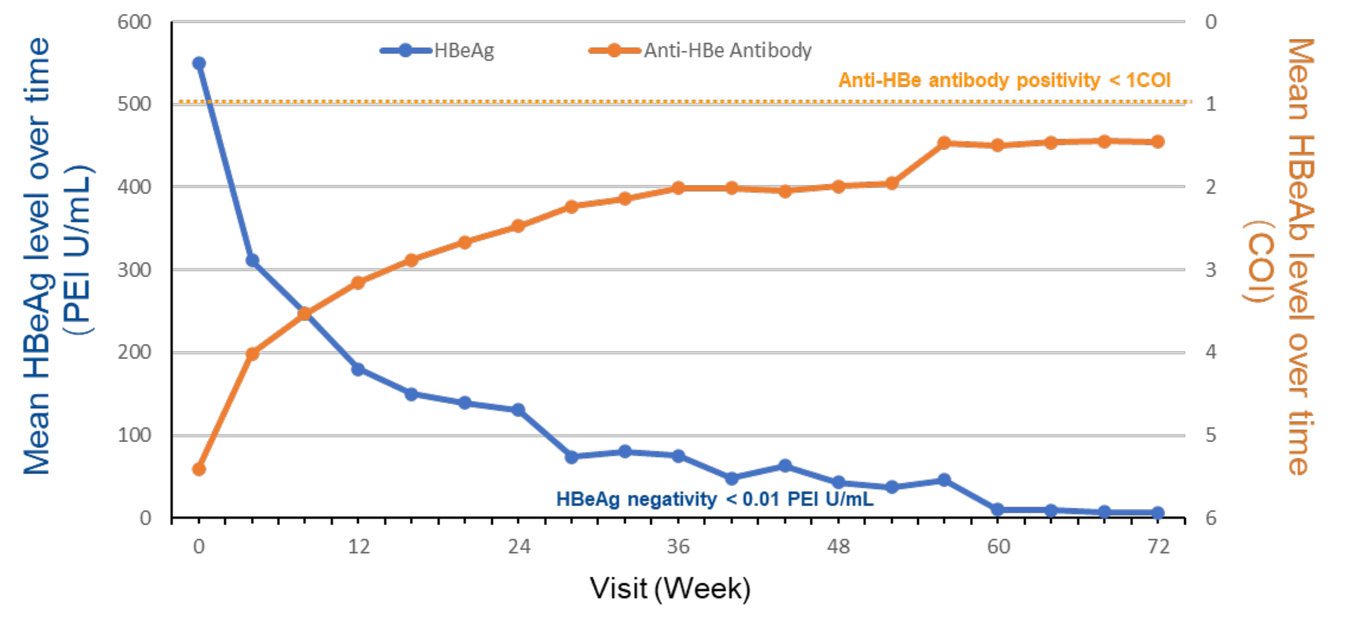
HBeAg
Anti-HBe Antibody
Anti-HBe antibody positivity < 1COI
HBeAg negativity < 0.01 PEI U/mL
Mean HBeAg level over time (PEI U/mL)
Mean HBeAb level over time (COI)
Visit (Week)

Conclusions

- Untreated HBeAg+ CHB subjects given 300 mg of ALG-000184 ± ETV for up to 72 weeks resulted in:
 - A favorable safety profile
 - HBV DNA suppression (< LLOQ10 IU/mL) rates by Week 48 and 72 were 60% and 90%, respectively for subjects receiving 300 mg ALG-000184 monotherapy and 33% and 50%, respectively for subjects receiving 300 mg ALG-000184 + ETV
 - Longer time to achieve HBV DNA suppression is required in subjects with higher baseline HBV DNA
 - All subjects achieved HBV RNA suppression by Week 40
 - No viral breakthrough was observed in ALG-000184 monotherapy up to 72 weeks
 - Multi-$\log_{10}$ declines in HBsAg, HBeAg and HBcrAg were observed
 - HBsAg levels did not further decline beyond Week 48 indicating that integrated HBV DNA maybe the contributing source for HBsAg
- ALG-000184 monotherapy achieves sustained HBV DNA suppression levels by Week 48 which appear to exceed those reported by nucleo(t)sides [7], suggesting the possibility for superior chronic suppression treatment for CHB patients.
- Based on its reduction of antigen levels, ALG-000184 may also play a pivotal role in combination with complementary MOAs for functional cure.
- Further studies are warranted and Phase 2 enabling activities for ALG-000184 are underway.

References

1. AASLD, EASL, APASL guideline; 2. Q Ling EASL 2020; 3. MF Yuen APASL Oral 2024; 4. Ed Gane APASL Oral 2023; 5. JL Hou APASL Oral 2023; 6. JL Hou APASL Oral 2024; 7.Buti M. et. al, Lancet 2016

Acknowledgement

The authors wish to thank the subjects for participating in this clinical study. The Sponsor is grateful to the staff of the clinical sites and to NOVOTECH, KINGMED and TIGERMED for assisting in the conduct of the study. The authors also wish to thank Aligos team members Jen Rito and Chris Burnett for their aid in the conduct of the study.