Contact Information: Stanley Wang (swang@aligos.com)

ALIGOS THERAPEUTICS

Abstract #03069

Safety and pharmacokinetics of single and multiple ascending doses of ALG-097558, a pan-coronavirus protease inhibitor, in healthy volunteers

Authors: Denisa Wilkes[1], Christina Yeung[1], Karen Dhillon[1], Kha Le[2], Stanley Wang[2], Chris Burnett[2], Jen Rito[2], Kusum Gupta[2], Doug Clark[2], Andreas Jekle[2], Sandra Chang[2], Naqvi Mohammed[2], Qiaoying Li[2], Meenakshi Venkatraman[2], Leonid Beigelman[2], Lawrence Blatt[2], Tse-I Lin[2], Sushmita Chanda[2], Matthew McClure[2]

1 – Hammersmith Medicines Research (HMR), United Kingdom; 2 – Aligos Therapeutics, Inc., United States

Disclosures: DW, CY, KD: none; KL, SW, CB, JR, KG, DC, AJ, SC, NM, QL, MV, LB, LB, TL, SC, MM: Aligos employees

INTRODUCTION

The highly transmissible viral infection COVID-19, caused by the novel coronavirus SARS-CoV-2 and its emerging (sub)variants, remains a major public health concern. Current standard of care therapeutics for COVID-19, including Paxlovid™ (nirmatrelvir/ritonavir), are limited by a combination of suboptimal potency and significant contraindications due to risk of drug-drug interactions (DDIs). Therefore, a significant unmet need remains for more potent oral treatments for acute SARS-CoV-2 infection that are not limited due to DDI risk with co-administered drugs. There is additionally an urgent need for antiviral treatments with pan-coronavirus activity to prepare for future pandemics.

BACKGROUND

ALG-097558 (ALG-558), which was discovered collaboratively by Aligos Therapeutics, KU Leuven, Cistim and CD3, is a novel small molecule inhibitor of SARS-CoV-2 3CLpro with potent antiviral activity (≥6-fold more active than nirmatrelvir in vitro), pan-coronavirus activity (including SARS-CoV-2, MERS-CoV and SARS-CoV-1), and a high barrier to resistance that is being developed as a ritonavir-free COVID-19 treatment and for future coronavirus pandemics.[1] Here we report preliminary results from Parts 1 (unblinded data), 2 (blinded data) and 6 (open label) of the ongoing multi-part first-in-human Phase 1 study, ALG-097558-701 (NCT05840952), which is being conducted at a single clinical pharmacology unit (HMR) in the United Kingdom.

METHODS

- Part 1 (single ascending dose; SAD) and Part 2 (multiple ascending dose; MAD) were double-blind, randomized, placebo (PBO)-controlled studies (Figure 1) evaluating the safety, tolerability and pharmacokinetics (PK) of single or multiple (twice daily (BID) for 7 days) oral doses, respectively, of ALG-097558/PBO in healthy volunteers (HV).
- For each Part 1 and 2 cohort, 8 HVs were planned for randomization to ALG-097558 or PBO in a 3:1 ratio, except in SAD Cohort 6 where 11 HVs randomized 9:2 was planned. SAD Cohort 2 (200 mg) subjects received a first dose of study drug fasted followed by a second dose fed (high-fat/high-calorie meal) after 3 days of washout, to assess the effect of food on the PK profile. Study drug was delivered as a solution formulation (containing polyethylene glycol).
- Part 6 was an open-label, fixed sequence, crossover study in 12 HVs assessing the relative bioavailability (BA) in plasma of a tablet vs. solution formulation of ALG-097558 (600 mg dose) and food effect of the tablet formulation (Figure 1).
- Safety assessments (adverse events (AEs), vital signs, electrocardiograms (ECG) and laboratories) and plasma/urine PK samples were routinely collected and analysed.
- Plasma and urine concentrations of ALG-097558 and its major metabolite, ALG-097730, were quantified using validated liquid chromatography-tandem mass spectrometry methods.

Figure 1: Study Designs

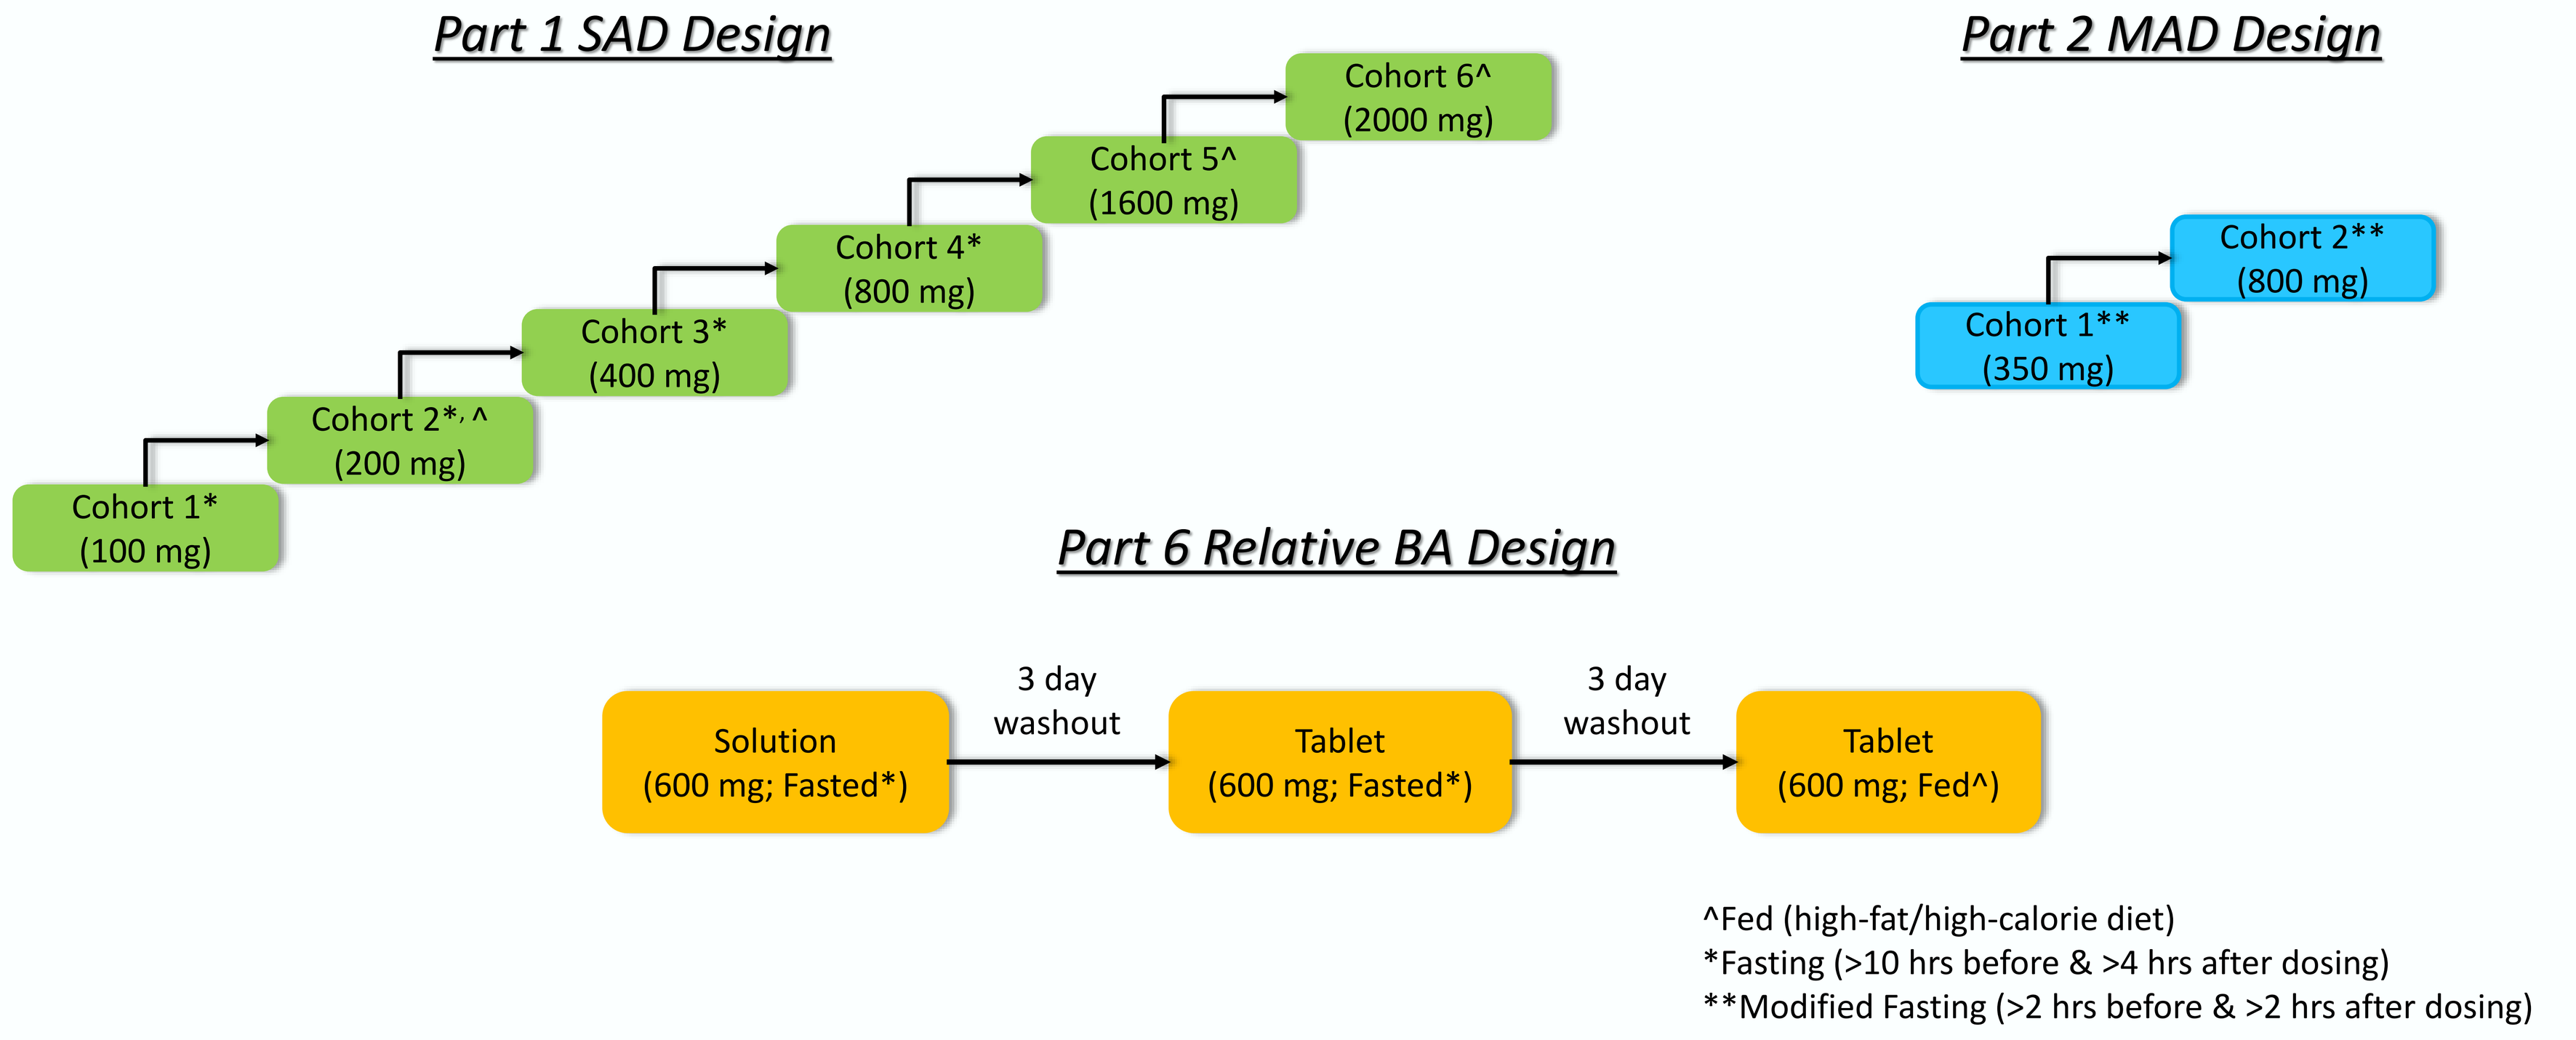
Part 1 SAD Design
Cohort 1* (100 mg)
Cohort 2*,^ (200 mg)
Cohort 3* (400 mg)
Cohort 4* (800 mg)
Cohort 5^ (1600 mg)
Cohort 6^ (2000 mg)
Part 2 MAD Design
Cohort 1** (350 mg)
Cohort 2** (800 mg)
Part 6 Relative BA Design
Solution (600 mg; Fasted*)
3 day washout
Tablet (600 mg; Fasted*)
3 day washout
Tablet (600 mg; Fed^)
^Fed (high-fat/high-calorie diet)
*Fasting (>10 hrs before & >4 hrs after dosing)
**Modified Fasting (>2 hrs before & >2 hrs after dosing)

RESULTS – BASELINE DEMOGRAPHICS

- In both Part 1 (SAD) and Part 2 (MAD), the baseline characteristics were generally similar across cohorts/parts (Table 1) and typical for a HV population
- In Part 6 (N = 12), 100% of subjects were male and mostly white race (75%), with a mean (SD) age of 35.8 (2.4) years and mean (SD) BMI of 24.7 (1.1) kg/m^2

Table 1: SAD and MAD Baseline Demographics

	SAD							MAD (ALG-097558/Placebo)	
	100 mg (N = 6)	200 mg (N = 6)	400 mg (N = 6)	800 mg (N = 6)	1600 mg (N = 6)	2000 mg (N = 9)	Placebo (N=12)	350 mg (N = 8)	800 mg (N = 7)
Age, years (mean (SD))	31.0 (2.6)	34.7 (5.3)	34.7 (4.3)	36.7 (4.0)	40.7 (4.1)	37.4 (4.0)	37.5 (2.6)	34.4 (3.7)	31.7 (2.4)
% Male	100%	100%	100%	100%	100%	100%	100%	100%	100%
Race									
White	4 (67%)	3 (50%)	3 (50%)	6 (100%)	5 (83%)	8 (89%)	6 (50%)	4 (50%)	3 (42%)
Black	0 (0%)	2 (33%)	2 (33%)	0 (0%)	0 (0%)	1 (11%)	2 (17%)	2 (25%)	2 (29%)
Other	2 (33%)	1 (17%)	1 (17%)	0 (0%)	1 (17%)	0 (0%)	4 (33%)	2 (25%)	2 (29%)
BMI, kg/m^2	23.8 (0.7)	26.8 (1.2)	26.4 (0.7)	23.9 (2.1)	26.3 (1.3)	23.6 (0.6)	24.8 (1.0)	24.6 (0.7)	23.7 (1.2)

REFERENCES: 1. Jekle A et al., 7th ISIRV-AVG Conference (May 2023). Abstract AASU0006. 2. Owen DR et al. Science. 2021; 374:1586-1593. 3. Jekle A et al. RespiDart 2022 Conference.

RESULTS – SAFETY

- No serious or treatment emergent AEs (TEAEs) leading to early discontinuation occurred
- All TEAEs were mild (Grade 1) or moderate (Grade 2) in severity
- The most common (≥2 subjects) TEAEs were:
 - Part 1 SAD – headache (PBO (N=1); 400 mg ALG-558 (N=1); 1600 mg ALG-558 (N=1))
 - Part 2 MAD – loose stools (N=3 in 800 mg PBO/ALG-558 cohort)
 - Part 6 Relative BA – no TEAE reported in more than 1 subject
- All treatment-emergent laboratory abnormalities were Grade 1 except for:
 - SAD 200 mg fed period: transient, asymptomatic Grade 2 lipase elevation (N=1 ALG-558) as well as Grade 2 cholesterol (N=2 ALG-558), LDL (N=1 ALG-558; N=1 PBO) and triglyceride (N=1 ALG-558) elevations in the context of a high-fat/high-calorie diet and/or baseline Grade 1 cholesterol or LDL elevations
 - Grade 2 cholesterol (N=1 SAD 800 mg ALG-558; N=1 PBO; N=1 MAD 350 mg PBO/ALG-558) and LDL (N=1 SAD 800 mg ALG-558) elevations in subjects with baseline Grade 1 cholesterol and/or LDL elevations
 - Grade 3 low hemoglobin that was asymptomatic and transient in 1 subject (SAD 800 mg ALG-558) with a baseline Grade 1 low hemoglobin and a medical history of mild anemia
- No clinically concerning ECG, vital sign or physical examination findings were reported

RESULTS – PHARMACOKINETICS

- There was a dose-related increase in plasma exposure with rapid absorption, low-moderate PK variability (doses ≥200 mg), mean terminal $t_{1/2}$ ~2-9 hours (Figure 2) and minimal impact of high-fat/high-calorie diet on PK (Figure 3)
- Metabolite ALG-097330 exposure was ~28-41% of parent ALG-097558
- Urine excretion of ALG-097558 was low (<7% and 3% of total dose for parent and metabolite, respectively, at 1600 mg)
- MAD: There were dose-related increases in ALG-097558 plasma exposures (Day 1) with no accumulation. Decreases in plasma exposures were observed at 800 mg Q12H on Day 7, but steady-state C_{trough} levels on Days 1-5 remained >5 fold above serum-shifted antiviral $EC_{99.9}$
- Geometric mean ratio for C_{max}, AUC_{last}, and AUC_{inf} were 102%, 158%, and 155% for tablet vs. solution formulations, respectively, with no impact of food on tablet exposures

Figure 2: SAD ALG-097558 PK

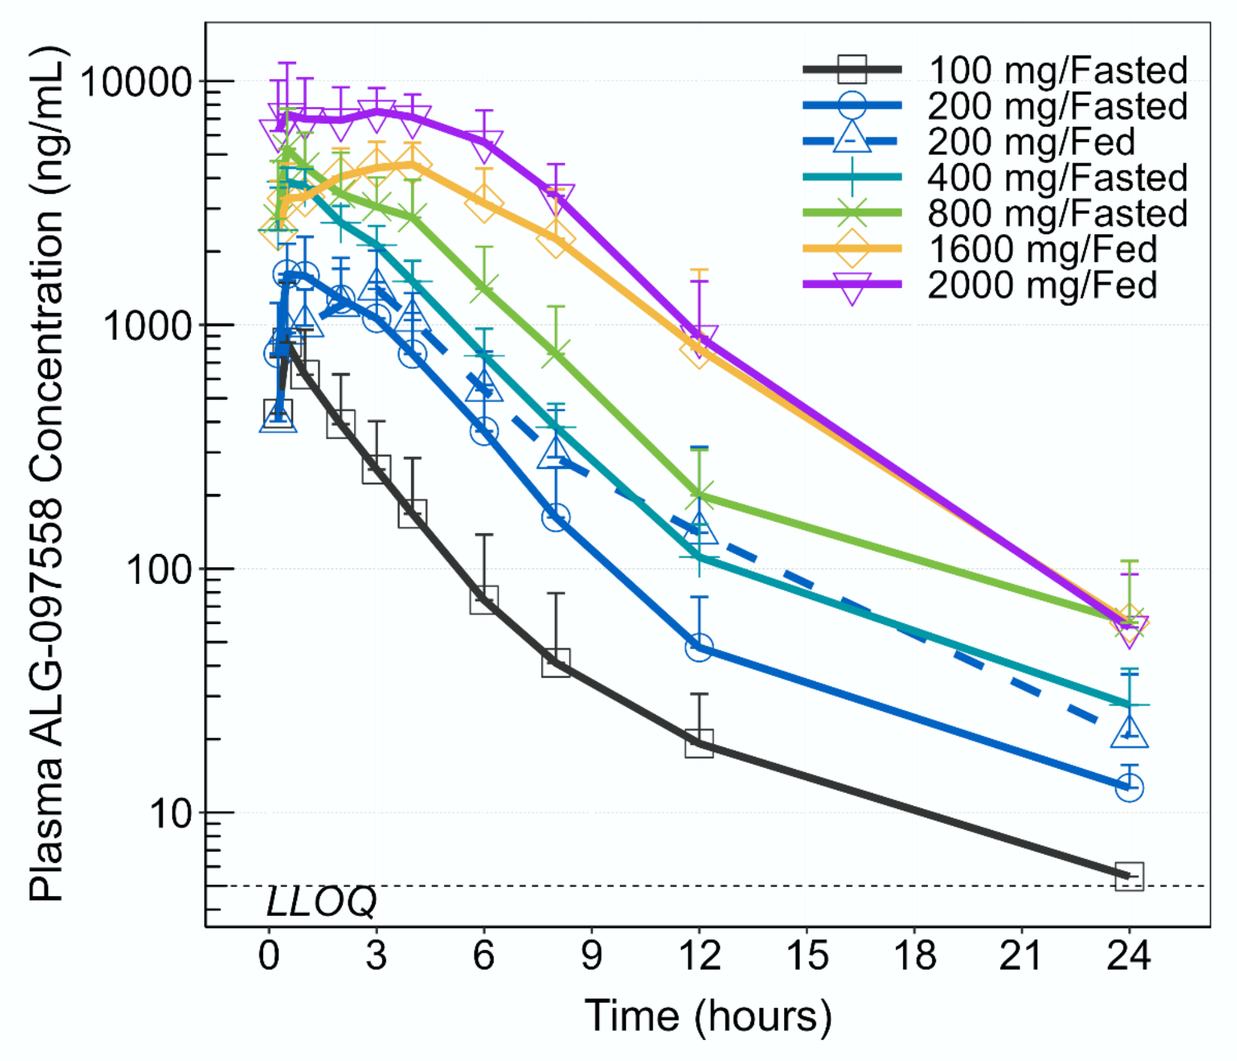
Plasma ALG-097558 Concentration (ng/mL)
10000
1000
100
10
LLOQ
0 3 6 9 12 15 18 21 24
Time (hours)
100 mg/Fasted
200 mg/Fasted
200 mg/Fed
400 mg/Fasted
800 mg/Fasted
1600 mg/Fed
2000 mg/Fed

Figure 3: Relative Bioavailability/Food Effect

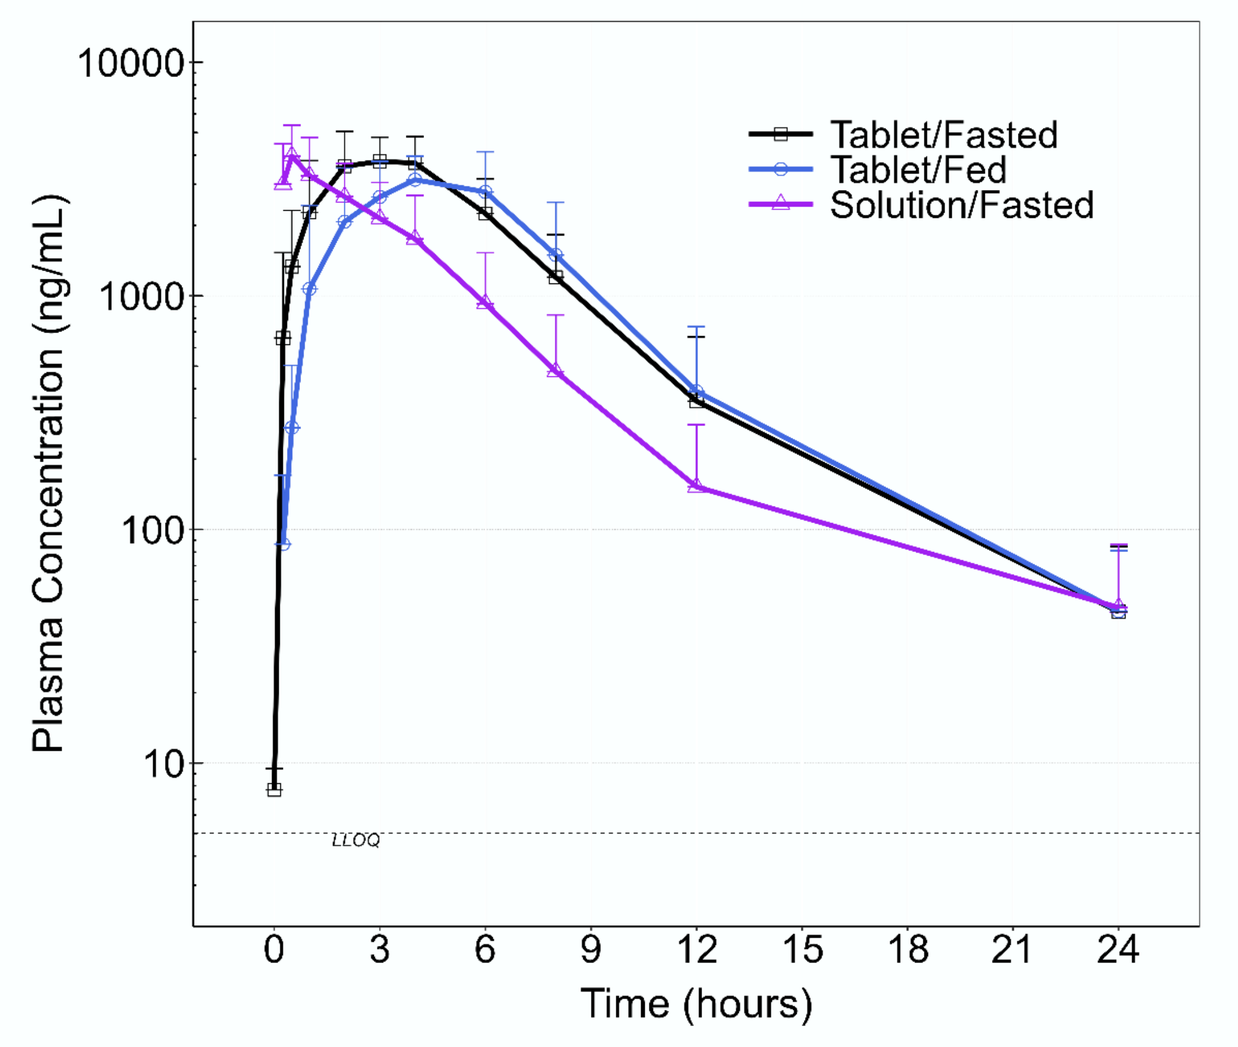
Plasma Concentration (ng/mL)
10000
1000
100
10
0 3 6 9 12 15 18 21 24
Time (hours)
Tablet/Fasted
Tablet/Fed
Solution/Fasted

Table 2: ALG-097558 PK Parameters for SAD, MAD and Relative BA/Food Effect

	SAD							MAD		Relative BA/Food Effect		
	100 mg Fasted	200 mg Fasted	200 mg Fed	400 mg Fasted	800 mg Fasted	1600 mg Fed	2000 mg Fed	350 mg Modified Fast	800 mg Modified Fast	600 mg Solution Fasted	600 mg Tablet Fasted	600 mg Tablet Fed
C_{max} (ng/mL)	753 (69.6)	1680 (30.9)	1490 (33.8)	4150 (12.2)	5140 (39.7)	4610 (18.9)	8970 (36.8)	3050 (26)	3310 (51.4)	4180 (37.2)	4150 (26.1)	3780 (23.3)
AUC_{0-12} (ng.hr/mL)	1750 (63.4)	6190 (40.6)	7000 (37.6)	14300 (13.3)	20000 (41.8)	32600 (26.4)	54800 (27.4)	10100 (50.2)	13100 (37.4)	14200 (45.7)	22500 (30.6)	19800 (29.1)
$AUC_{0-\infty}$ (ng.hr/mL)	1780 (64.4)	6460 (41.6)	7650 (44.1)	15500 (13.7)	22000 (44.4)	35800 (28.7)	58700 (29.6)	11200 (58.4)	14300 (40.6)	15900 (48.4)	24300 (34.7)	21700 (34)
t_{max} (hr)	0.5 (0.5,1)	0.75 (0.5,3)	2.5 (0.5,4)	0.75 (0.5,1)	0.5 (0.5,3)	3.5 (2,8)	2 (0.25,6)	0.5 (0.25,1)	1 (0.5,1)	0.5 (0.25,1)	3.5 (2,6)	4 (1,6)
$t_{1/2}$ (hr)	1.95 (36.5)	3.26 (34.2)	4.03 (24.8)	7.4 (75.6)	9.19 (43.3)	2.96 (9.96)	2.99 (35.3)	2.37 (9.25)	6.13 (33.7)	9.99 (53.1)	3.99 (36.5)	3.65 (32.3)

Values shown are geometric mean (geometric CV) except for t_{max} as median (minimum, maximum) and $t_{1/2}$ as arithmetic mean (CV)

CONCLUSIONS

- Single (up to 2000 mg) and multiple (up to 800 mg BID for 7 days) doses of the potent, pan-coronavirus protease inhibitor ALG-097558 were well tolerated in healthy volunteers.
- The PK profile of ALG-097558 supports twice daily ritonavir-free dosing without food effect.
- The projected efficacious dose range for the ALG-097558 tablet formulation for future clinical studies in COVID-19 patients is 200–600 mg BID x 5 days (without ritonavir). These regimens are estimated to maintain C_{trough} levels that are up to 9.5-fold above the antiviral $EC_{99.9}$, which exceeds the ~6-fold antiviral $EC_{99.9}$ C_{trough} that are maintained by Paxlovid.[2,3]

ACKNOWLDGEMENTS: The authors wish to thank the subjects participating in this clinical study. The Sponsor is grateful to the staff at HMR.

34th ECCMID EUROPEAN CONGRESS OF CLINICAL MICROBIOLOGY AND INFECTIOUS DISEASES

Barcelona, Spain 27 – 30 April 2024