DocuSign Envelope ID: EA651055-E866-49C2-ACF0-53D11E1ED3D9

AASLD The Liver Meeting® Nov. 10-14, 2023

PROLONGED (>24 WEEK) DOSING WITH THE ORAL CAM-E COMPOUND ALG-000184 RESULTS IN MULTI-LOG REDUCTIONS IN HEPATITIS B SURFACE ANTIGEN, HBV DNA, AND HBV RNA LEVELS IN UNTREATED E ANTIGEN POSTIVE SUBJECTS WITH CHRONIC HEPATITIS B

HOU JinLin [1]; DING Yanhua [2]; LIANG Xieer [1]; XU Jia [2]; YUEN Man-Fung [3]; GANE Ed [4]; AGARWAL Kosh [5]; NIU Junqi [2]; WU Min [6]; LE Kha [6]; WESTLAND Christopher [6]; MADERAZO Maida [6]; HARRINGTON Genevieve [6]; BLATT Lawrence M [6]; BEIGELMAN Leonid N. [6]; CHANDA Sushmita [6]; LIN Tse-I [6]; McCLURE Matthew [6]; FRY John [6]

1. *Nanfang Hospital of Southern Medical University. 2. The First Hospital of Jilin University. 3. Department of Medicine, School of Clinical Medicine, The University of Hong Kong. 4. Faculty of Medicine, University of Auckland. 5. Institute of Liver Studies, King's College Hospital. 6. Aligos Therapeutics, Inc.*

ALIGOS THERAPEUTICS

Abstract #41220
Poster #1483-C

INTRODUCTION

- The aims of treatment of chronic hepatitis B (CHB) are to achieve sustained suppression of HBV replication and remission of liver disease. [1]
- Achieving this goal is challenging due to persistence of covalently closed circular DNA (cccDNA), the transcriptional template of HBV, in the nucleus of hepatocytes. [1]
- In vitro, ALG-000184 has been shown to inhibit HBsAg production through blocking intrahepatic cccDNA establishment (2nd mechanism of action [MOA]) in addition to inhibiting viral replication through modulating pg-RNA encapsidation (1st MOA).[2]
- Multi-log reductions in viral load and HBsAg have been reported previously in untreated HBeAg+ subjects receiving 300 mg ALG-000184 + entecavir (ETV) x≤36 weeks.[3]
- Here we report emerging data in untreated HBeAg+ CHB subjects receiving 300 mg ALG-000184 + ETV x ≤48 weeks.

AIM

To evaluate the safety, pharmacokinetics (PK), and antiviral activity of multiple doses of ALG-000184 in CHB subjects

MATERIALS AND METHODS

ALG-000184-201 (NCT04536337) is a multipart, double blind, randomized placebo-controlled Phase 1 study. Data from Parts 1-3 and Part 4 Cohort 1, which are all complete, have been previously reported. [3,4] Part 4 Cohort 2 is an ongoing double blind, randomized (4 active:1 placebo) cohort that is evaluating oral daily doses of 300 mg ALG-000184 or placebo in combination with 0.5 mg ETV for 12 weeks in untreated HBeAg+ CHB subjects. After dosing for 12 weeks, all subjects subsequently receive 300 mg ALG-000184 + ETV for a total planned treatment duration of 96 weeks (Figure 1).

Figure 1 Design of Part 4 Cohort 2 in Study ALG-000184-201

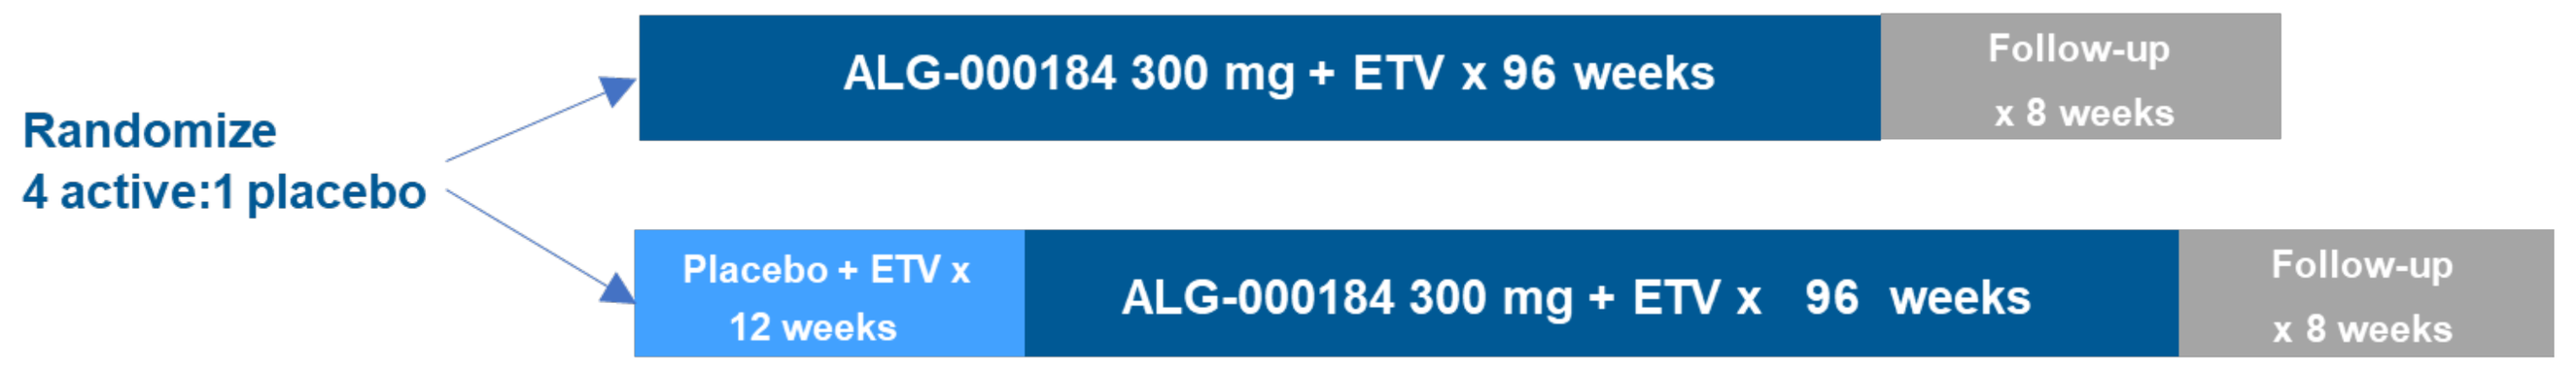
Randomize
4 active:1 placebo
ALG-000184 300 mg + ETV x 96 weeks
Follow-up x 8 weeks
Placebo + ETV x 12 weeks
ALG-000184 300 mg + ETV x 96 weeks
Follow-up x 8 weeks

Throughout the study, safety assessments (adverse events [AEs], vital signs, electrocardiogram [ECG] and laboratories), PK, and viral markers are regularly collected. A Study Review Committee (SRC) and ALT Flare Committee (AFC) review safety and PK data on a regular basis for study oversight and to determine dosing regimen, including total dosing duration. Virology assays for this cohort have the following cutoffs:

- HBV DNA: Roche Cobas® assay
 - Lower Limit Quantification (LLOQ): 10 IU/mL
 - Lower Limit Detection (LLOD): 3.5 IU/mL
- HBV RNA: (China local assay)
 - LLOQ: 200 copies/mL; LLOD: 100 copies/mL
- HBsAg: Roche Elecsys® HBsAg II quant II
 - LLOQ: 0.05 IU/mL

Antiviral activity data are summarized here as change from baseline and include subjects on study drug (confirmed by PK) at the relevant timepoint.

RESULTS

BASELINE CHARACTERISTICS

- 15 subjects were enrolled. All were Asian, HBV genotype B or C and HBeAg+
- Baseline characteristics were comparable between ALG-000184 (n=12) and placebo (n=3) arms
- 47% (7/15) subjects had normal ALT at screening and baseline.
- 3 of 15 enrolled subjects discontinued prematurely due to non-safety related personal reasons (N=2) and confirmed non-compliance beginning at Week 12 (N=1).

Table 1: Baseline characteristics

Part 4 Cohort 2 300 mg ALG-000184/Placebo + ETV (N=15)	184+ETV (n=12)	PLB+ETV (n=3)
Age, years, mean (SEM)	32.3 (3.0)	28 (2.6)
Male, n(%)	6 (50)	2 (67)
Asian, n(%)	12 (100)	3 (100)
BMI, kg/m^2, mean (SEM)	21.2 (0.9)	22.3 (1.6)
HBV Genotype, n(%)	B: 4 (33) C: 8 (67)	B: 1 (33) C: 2 (67)
HBV DNA $\log_{10}$ IU/mL, mean (SEM)	8.1 (0.2)	7.8 (0.6)
HBV RNA $\log_{10}$ copies/mL, mean (SEM)	6.7 (0.3)	6.5 (0.5)
HBsAg $\log_{10}$ IU/mL, mean (SEM)	4.5 (0.2)	4.1 (0.2)
ALT U/L, mean (SEM)	41.2 (6.6)	38.7 (5.2)

PLB: Placebo; SEM: standard error of mean; wks: weeks

SAFETY

- 300 mg ALG-000184 + 0.5 mg ETV for up to 48 weeks was generally well tolerated.

Table 2: Summary of Treatment Emergent AEs

Part 4 Cohort 2 300 mg ALG-000184/PLB + ETV (N=15)	184+ETV (n=12)	PLB+ETV (n=3)
Serious Adverse Events (SAEs)	0	0
Treatment emergent AEs (TEAEs) leading to study drug discontinuation	0	0
Subjects with Grade ≥ 3 TEAEs	2 ↑ ALT/AST (n=2)*,** ↑ neutropenia (n=1)**	0
Concerning TEAE, laboratory, ECG, vital sign, or physical examination trends	None	

* One subject experienced a Grade 4 ALT elevation with associated Grade 2 AST elevations unlikely related to study drug on Day 171.
** Another subject experienced a Grade 4 ALT elevation with associated Grade 3 AST elevation possibly related to study drug on Day 41. The same subject experienced Grade 4 neutropenia probably related to an acute upper respiratory infection, per investigator. This event resolved following the resolution of this infection and despite continued dosing of study drug.

Neither transaminase event was associated with concerning symptoms or hepatic synthetic dysfunction and both events resolved despite continued dosing. The AFC assessed these events as not being due to drug toxicity.

ANTIVIRAL ACTIVITY: HBV DNA and HBV RNA

- Of the 11 compliant subjects that initially received 300mg ALG-000184+ETV ≤ 48 weeks:
 - Mean reduction (SEM) of HBV DNA at Week 12 was 5.1 (0.3) $\log_{10}$ IU/mL. DNA declines extended during longer treatment to 6.7 $\log_{10}$ IU/mL at Week 48. HBV DNA < LLOQ was achieved in 4 subjects.
- Of 3 subjects that initially received Placebo + ETV for 12 weeks and then received 300mg ALG-000184+ETV x ≤ 48 weeks:
 - Mean reduction (SEM) of HBV DNA at Week 12 was 3.8 (0.1) $\log_{10}$ IU/mL. Additional DNA declines were observed after adding 300 mg ALG-000184 at Week 12; the mean DNA reduction was 6.4 $\log_{10}$ IU/mL at Week 48 and was similar to that seen in subjects initially receiving 300 mg ALG-000184 + ETV. HBV DNA < LLOQ was achieved in 2 of 3 subjects.
- RNA: profound (3.5 $\log_{10}$ copies/mL) reductions were observed in the first 12 weeks for subjects receiving ALG-000184+ETV vs. minimal change in RNA levels among subjects receiving ETV x 12 weeks. All subjects receiving 300 mg ALG-000184 + ETV ultimately achieved HBV RNA < LLOQ.

Figure 1: HBV DNA, HBV RNA and HBsAg Change from Baseline

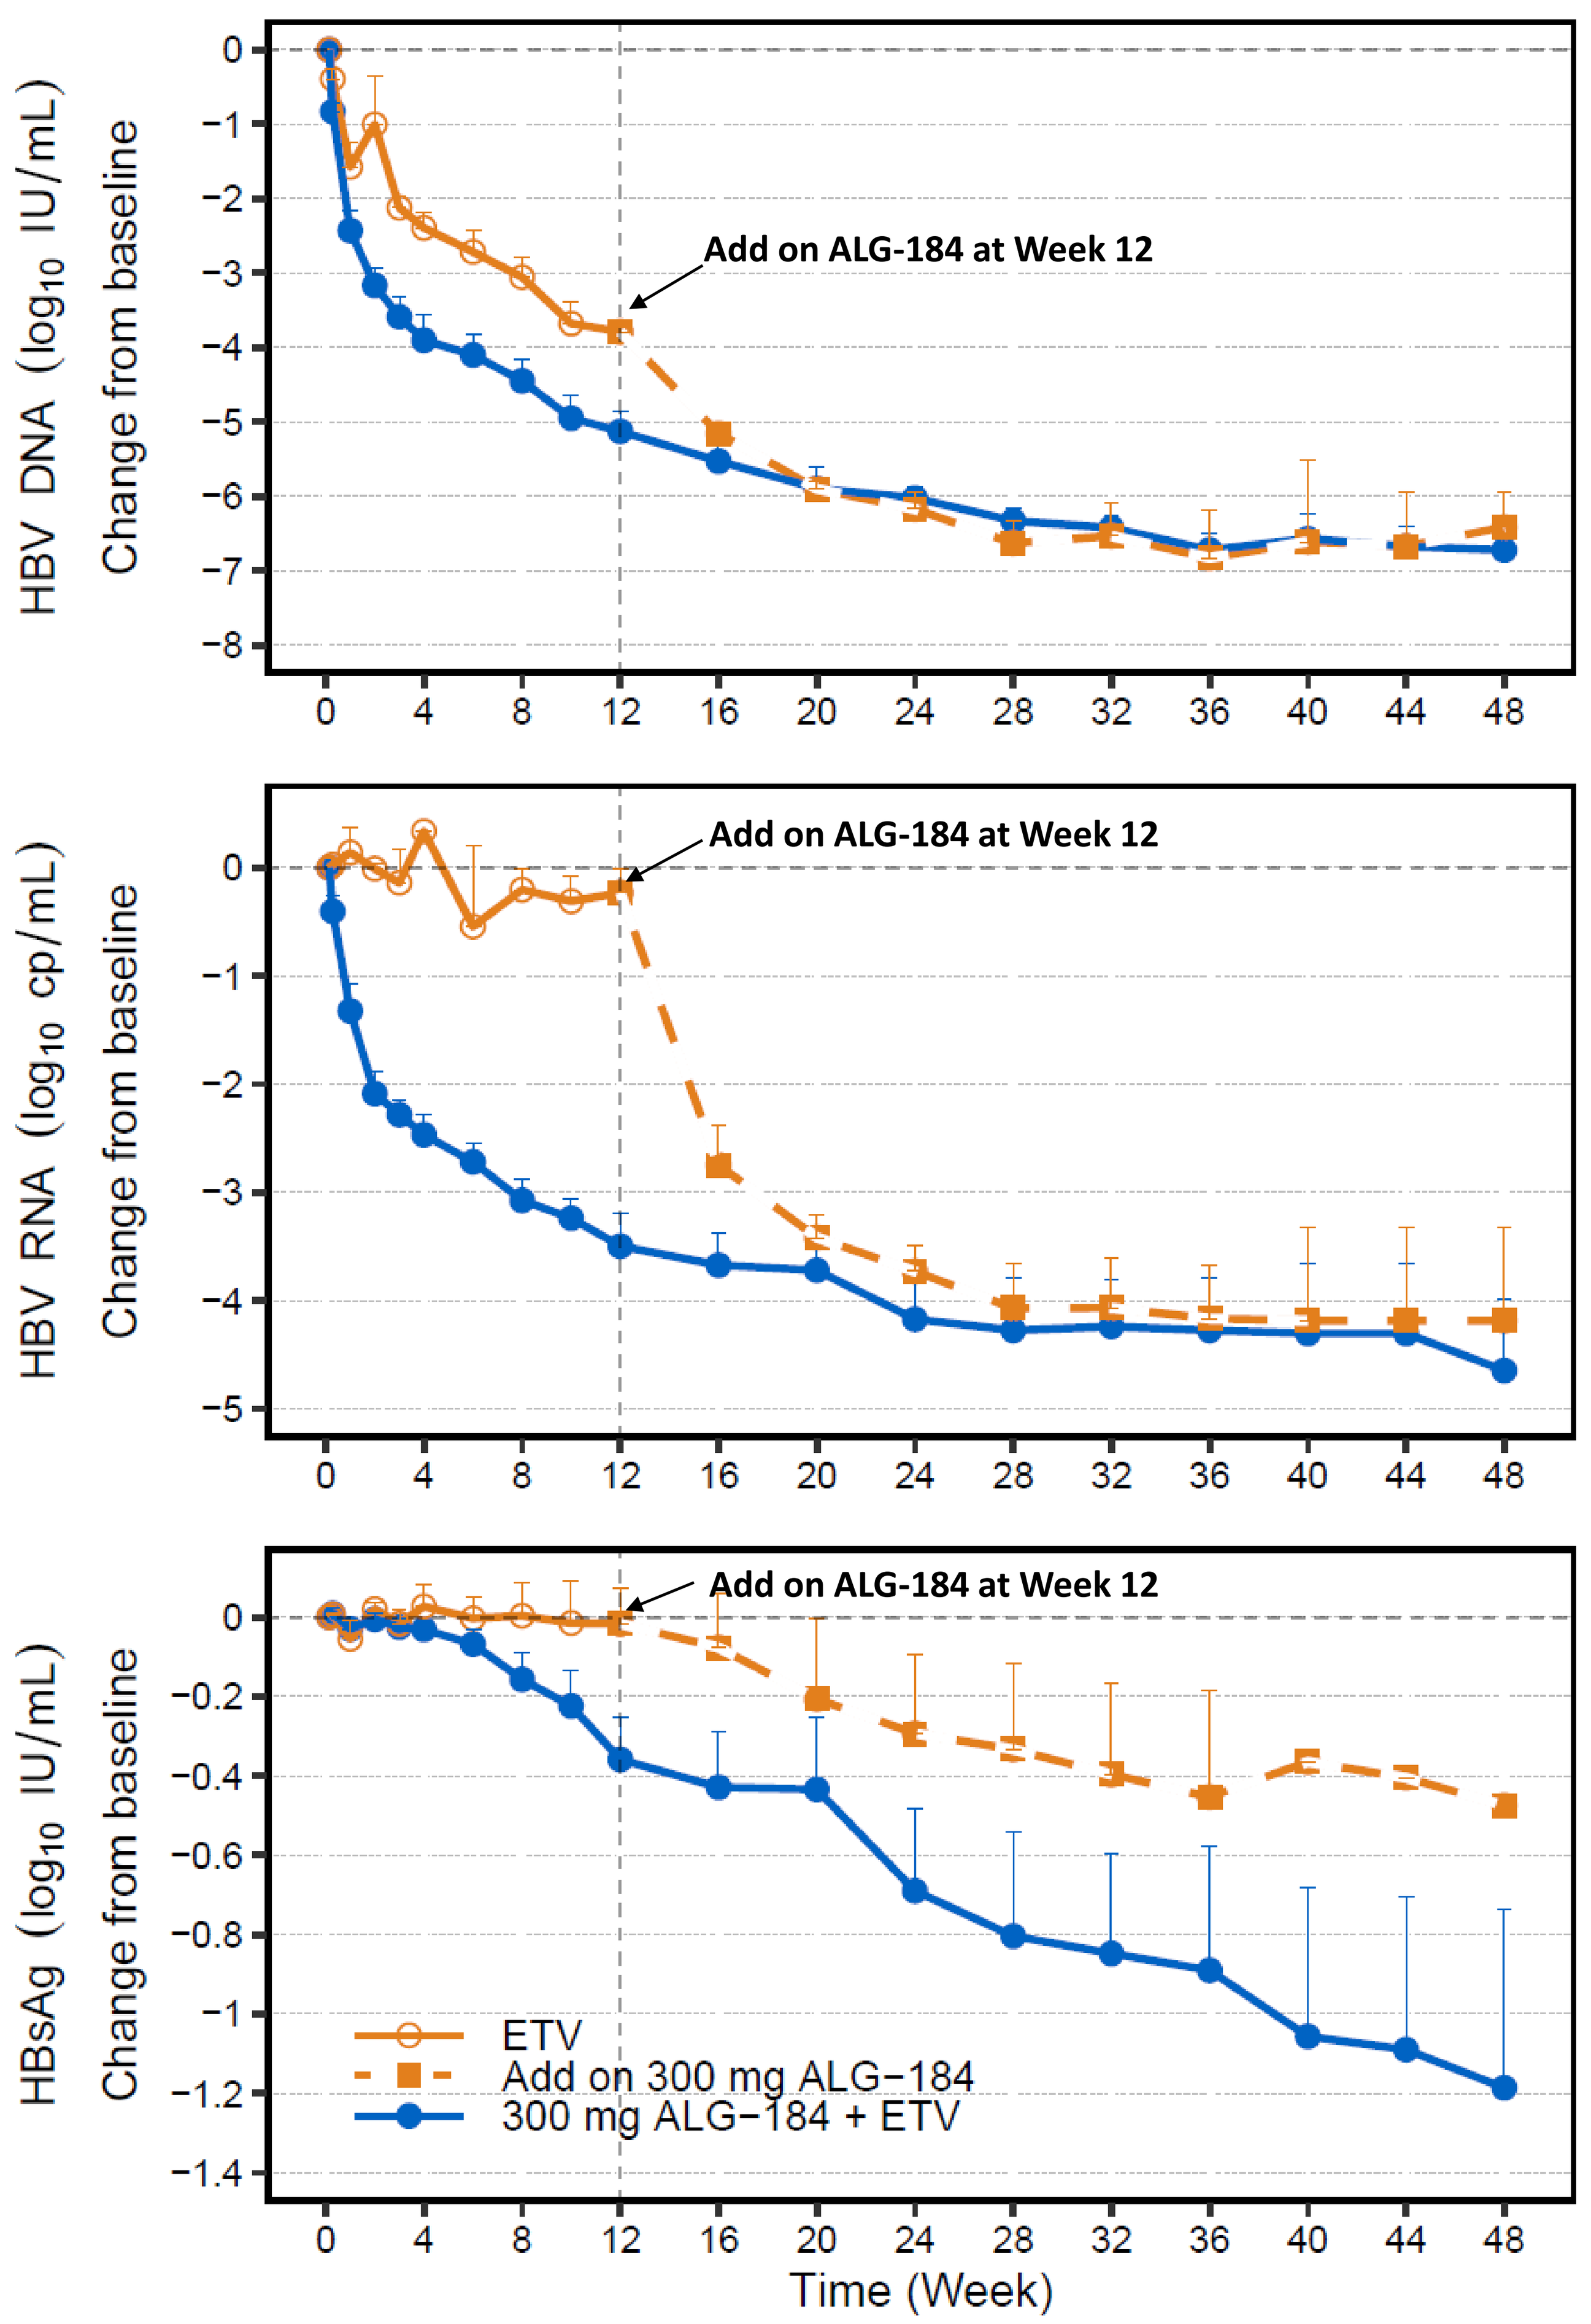
HBV DNA (log10 IU/mL) Change from baseline
Add on ALG-184 at Week 12
0
−1
−2
−3
−4
−5
−6
−7
−8
0 4 8 12 16 20 24 28 32 36 40 44 48
HBV RNA (log10 cp/mL) Change from baseline
Add on ALG-184 at Week 12
0
−1
−2
−3
−4
−5
0 4 8 12 16 20 24 28 32 36 40 44 48
HBsAg (log10 IU/mL) Change from baseline
Add on ALG-184 at Week 12
0
−0.2
−0.4
−0.6
−0.8
−1
−1.2
−1.4
ETV
Add on 300 mg ALG−184
300 mg ALG−184 + ETV
0 4 8 12 16 20 24 28 32 36 40 44 48
Time (Week)

ANTIVIRAL ACTIVITY: HBsAg

- Mean HBsAg levels declined by 0.4 $\log_{10}$ IU/mL and 1.2 $\log_{10}$ IU/mL after receiving 300 mg ALG-000184 + ETV for 12 and 48 weeks, respectively. The maximum reduction seen was 2.0 $\log_{10}$ IU/mL.
- No subject initially receiving placebo + ETV had any HBsAg change in the first 12 weeks of treatment. Subsequently, mean HBsAg levels declined by 0.5 $\log_{10}$ IU/mL at Week 48
- Similar HBsAg declines were observed in subjects regardless of baseline ALT level. The mean HBsAg reductions at Week 12, 24 and 48 in subjects with normal ALT were 0.3 (0.2), 0.8 (0.4) and 1.0 (0.5) $\log_{10}$ IU/mL, respectively with a maximum reduction of 1.8 $\log_{10}$ IU/mL.

CONCLUSIONS

- Oral daily dosing with 300 mg ALG-000184 + ETV for up to 48 weeks in untreated HBeAg + CHB subjects resulted in:
 - A favorable safety profile
 - Potent viral suppression of HBV DNA and RNA which was greater than seen with ETV alone
 - Substantial mean HBsAg declines of 1.2 $\log_{10}$ IU/mL at Week 48 with maximum observed HBsAg declines reaching 2.0 $\log_{10}$ IU/mL.
 - Treatment in this cohort is ongoing and will evaluate if further reductions in HBsAg can be achieved with longer dosing
 - These data indicate ALG-000184 is a potent CAM-E compound with broad anti-HBV effects, including inhibiting intrahepatic cccDNA. Please see late breaker poster #5028-C for additional data from this and other cohorts and further discussion of this topic
 - ALG-000184 has the potential to be a backbone therapy for chronic HBV suppression or functional cure

ACKNOWLEDGMENTS

The authors wish to thank the subjects for participating in this clinical study. The Sponsor is grateful to the staff of the clinical sites and to Novotech, Kingmed and Tigermed for assisting in the conduct of the study. The authors also wish to thank Aligos-team member Kim Steel for her aid in the conduct of the study.

REFERENCES

1. Norah A. Terrault, Hepatology 2015; 2. Q Zhang EASL 2020. 3. Jinlin Hou, EASL 2023; 4. Ed Gane APASL 2023

DISCLOSURES

Hou J.: Aligos, Assembly Biosciences, Ascletis, Ascentage Pharma, Bristol-Myers Squibb, GlaxoSmithKline, Gilead Sciences, Janssen, Roche, Huahuihealth, Qilu Pharma. **Ding Y.:** nothing to disclose. **Liang X.:** nothing to disclose**. Xu J.:** nothing to disclose**. Yuen MF**: AbbVie, Aligos Therapeutics, AiCuris, Antios Therapeutics, Arbutus Biopharma, Arrowhead Pharmaceuticals, Assembly Biosciences, Clear B Therapeutics, Dicerna Pharmaceuticals, Finch Therapeutics, Fujirebio Incorporation, GlaxoSmithKline, Gilead Sciences, Immunocore, Janssen, Roche, Sysmex Corporation, Tune Therapeutics, Vir Biotechnology and Visirna Therapeutics. **Gane E**: AbbVie, Abbott Diagnostics, Aligos, Arbutus, Arrowhead, Assembly, Avalia, Clear B Therapeutics, Dicerna, Enanta, Gilead Sciences, GlaxoSmithKline, Janssen, Merck, Roche and Vir Bio. **Agarwal K**: Abbott, Aligos, Arbutus, Assembly, BMI, BI, Gilead, Janssen, Immunocore, Roche, Sobi, Vir Bio. **Niu J**: nothing to disclose. **Wu M**, **Le K, Westland C, Maderazo M, Harrington G, Blatt L, Beigelman L, Chanda S, Lin T, McClure M**: Employees of Aligos Therapeutics Inc.

CONTACT INFORMATION

Min Wu (mwu@aligos.com)